Cargocytes™: A Novel Enucleated Cell Therapy Platform to Drive Anti-Tumor Immunity

Christina N. Alarcón[1,2,3,+], Huawei Wang[1,2,+], Bei Liu[1,2], Felicia Watson[1,2], Stephen Searles[1,3], Calvin Lee[1], Jack D. Bui[1], and Richard L. Klemke[1,2*]
[+] Co-first authors, [1]Department of Pathology, [2]Moores Cancer Center, [3]Biomedical Sciences Program, University of California, San Diego, La Jolla, California.
*Corresponding Author: Richard L. Klemke PhD, Department of Pathology, UC San Diego, School of Medicine, 9500 Gilman Drive #0612, La Jolla, CA 92093. 858-822-5610; rklemke@ucsd.edu

Abstract

Adoptive cell therapy has emerged as a powerful new tool to treat cancer and other diseases. However, the clinical utility of many cell-based therapies is limited by inadequate production of therapeutic factors, production of unwanted factors, unwanted engraftment into the tumor, and poor controllability resulting in poor pharmacokinetics. Genetic and bioengineering strategies can overcome many limitations, but modification of genomic DNA or the introduction of new genetic material into therapeutic cells raises substantial safety concerns, ultimately making FDA approval difficult. To address these limitations, we developed a novel platform for cell-based therapies in which <u>any</u> cell type, including immortalized cells, can be extensively engineered and loaded with therapeutic cargo, and then rendered safe for patient administration through removal of the nucleus. Enucleated therapeutic cells (Cargocytes™) retain many desired biologic functions (endogenous and engineered), such as viability up to 72 hours, retention of cell surface markers/proteins, secretion of bioactive molecules, robust *in vitro* and *in vivo* chemotaxis, and delivery of a wide range of cancer-fighting cargos. Using this technology, we demonstrate that Cargocytes derived from hTERT-immortalized mesenchymal stem cells (hTERT MSCs) and engineered to express therapeutic levels of IL-12 can potently activate an anti-tumor immune response in a preclinical mouse model of triple negative breast cancer. hTERT MSC-derived Cargocytes robustly delivered IL-12 to the tumor with definable pharmacokinetics, activated known IL-12 biomarkers, and induced infiltration of tumor-fighting immune cells that inhibited tumor growth and improved animal survival. In addition, co-administration of Cargocytes secreting IL-12 and anti-PD-1 antibodies further improved animal survival without notable adverse events. Collectively, our findings indicate that Cargocytes provide a new means to deliver powerful immunomodulatory biologics to malignant tumors in a safe and controllable manner. The genetic tractability and excellent safety profile of the Cargocyte platform enables bioengineering of a wide range of designer therapeutics derived from genetically modified cells, immortalized stem cells, and even cancer cells, each with improved clinically relevant functions.

Background

➢MSCs are heavily investigated cell-based therapies for a wide variety of diseases based on their secretory, anti-inflammatory, tumor-trophic, regenerative, or wound healing properties [1].

➢Current MSC therapies are limited by source heterogeneity, few engineering options, and safety issues such as tumor promotion [2].

➢To address these limitations, we created a novel cell-therapy platform in which <u>any</u> cell can be extensively engineered, and then enucleated to be made safe for clinical use. Following ultracentrifugation in Ficoll density gradients [3], these enucleated cells (cytoplasts) retain functional organelles, cytoskeletal scaffolding, migrational abilities, survive up to 3 days, survive cryohibernation and cryopreservation, and can be engineered or loaded with many different combinations of therapeutic cargo (called **<u>Cargocytes™</u>**).

➢Cell-based cancer therapies aiming to stimulate the endogenous immune system frequently attempt delivery of cytokines such as IL-12 to induce T cell and NK cell proliferation and activation and IFNγ production. However, systemic delivery of IL-12 can cause adverse affects (fever, cytokine storms, rarely death) that has limited the therapeutic application [4].

➢Intratumoral injections are a route to limit systemic toxicities while achieving local therapeutic effects.

➢Checkpoint inhibitors such as anti-PD-1 antibodies have been used alone or in conjunction with existing immunotherapies [5].

➢This project investigates the use of engineered cargocytes to secrete IL-12 at the site of syngeneic E0771 mouse breast cancer cells [6] in C57Bl/6 mice.

Figure 1. Cargocytes are transfectable with and secrete sufficient quantities of exogenous, functional IL-12 mRNA

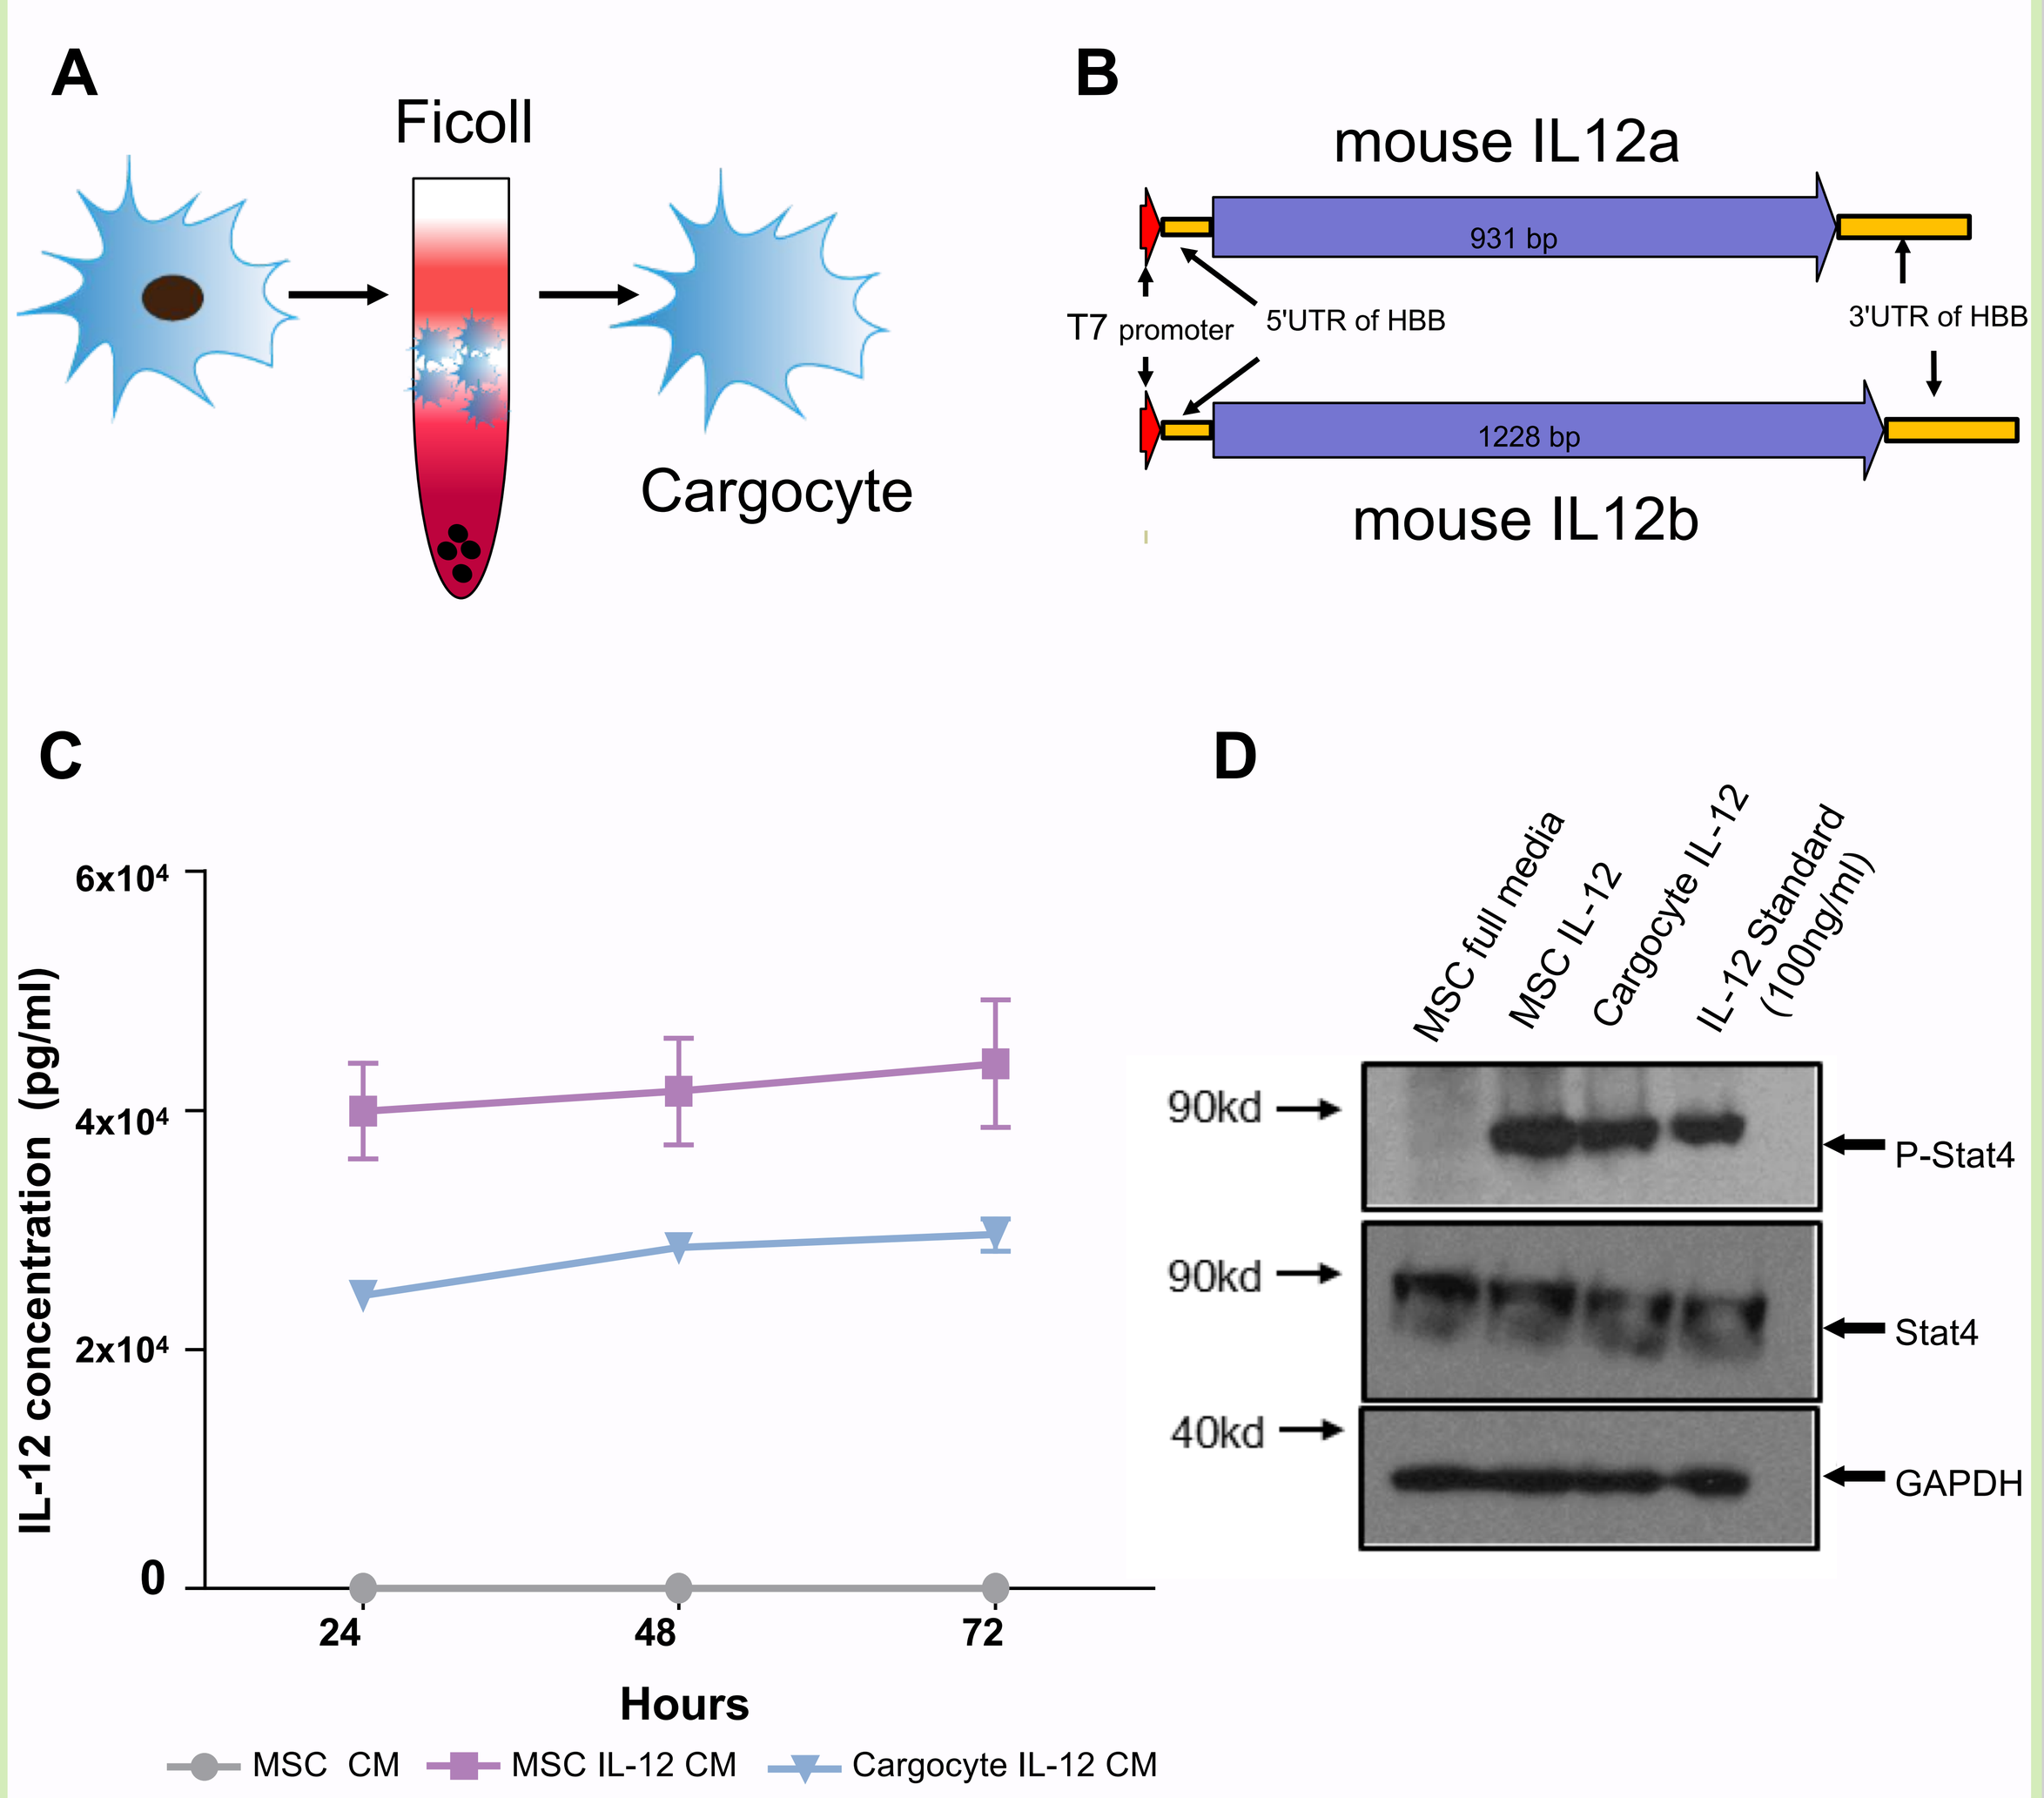
A
Ficoll
Cargocyte
B
mouse IL12a
931 bp
T7 promoter
5'UTR of HBB
3'UTR of HBB
1228 bp
mouse IL12b
C
6x10⁴
4x10⁴
2x10⁴
0
IL-12 concentration (pg/ml)
24
48
72
Hours
MSC CM
MSC IL-12 CM
Cargocyte IL-12 CM
D
MSC full media
MSC IL-12
Cargocyte IL-12
IL-12 Standard (100ng/ml)
90kd
90kd
40kd
P-Stat4
Stat4
GAPDH

Fig. 1. A) Schematic of cargocyte generation from any cell type by ultracentrifugation in Cytochalasin B-treated Ficoll gradients to extrude nuclei. B) Schematic of IL-12 mRNA construct (a and b strands) including T7 promoter, human beta globin (HBB) mRNA at the 5' and 3' UTR and pseudouridine modification for mRNA stability. C) MSCs or MSC-derived cargocytes were transfected with IL-12 mRNA in Lipofectamine suspension for 30 minutes, then plated at 2.5 x 10^4 cells/well of 24-well plate. Conditioned medium (CM) was collected at the indicated time points and the secreted IL-12 concentration determined by ELISA. MSC=non-transfected MSCs. MSC IL-12=MSC transfected with IL-12 mRNA. Cargocyte IL-12=cargocytes transfected with IL-12 mRNA. D) Serum-starved mouse splenocytes were treated for 30 minutes with either full media, purified IL-12 protein standard or the indicated CM collected from MSCs or cargocytes transfected with IL-12. The phosphorylated/activated form of Stat4 (P-Stat4) was determined by western blot.

Figure 2. Cargocytes secrete IL-12 in the tumor microenvironment with minimal amounts in blood

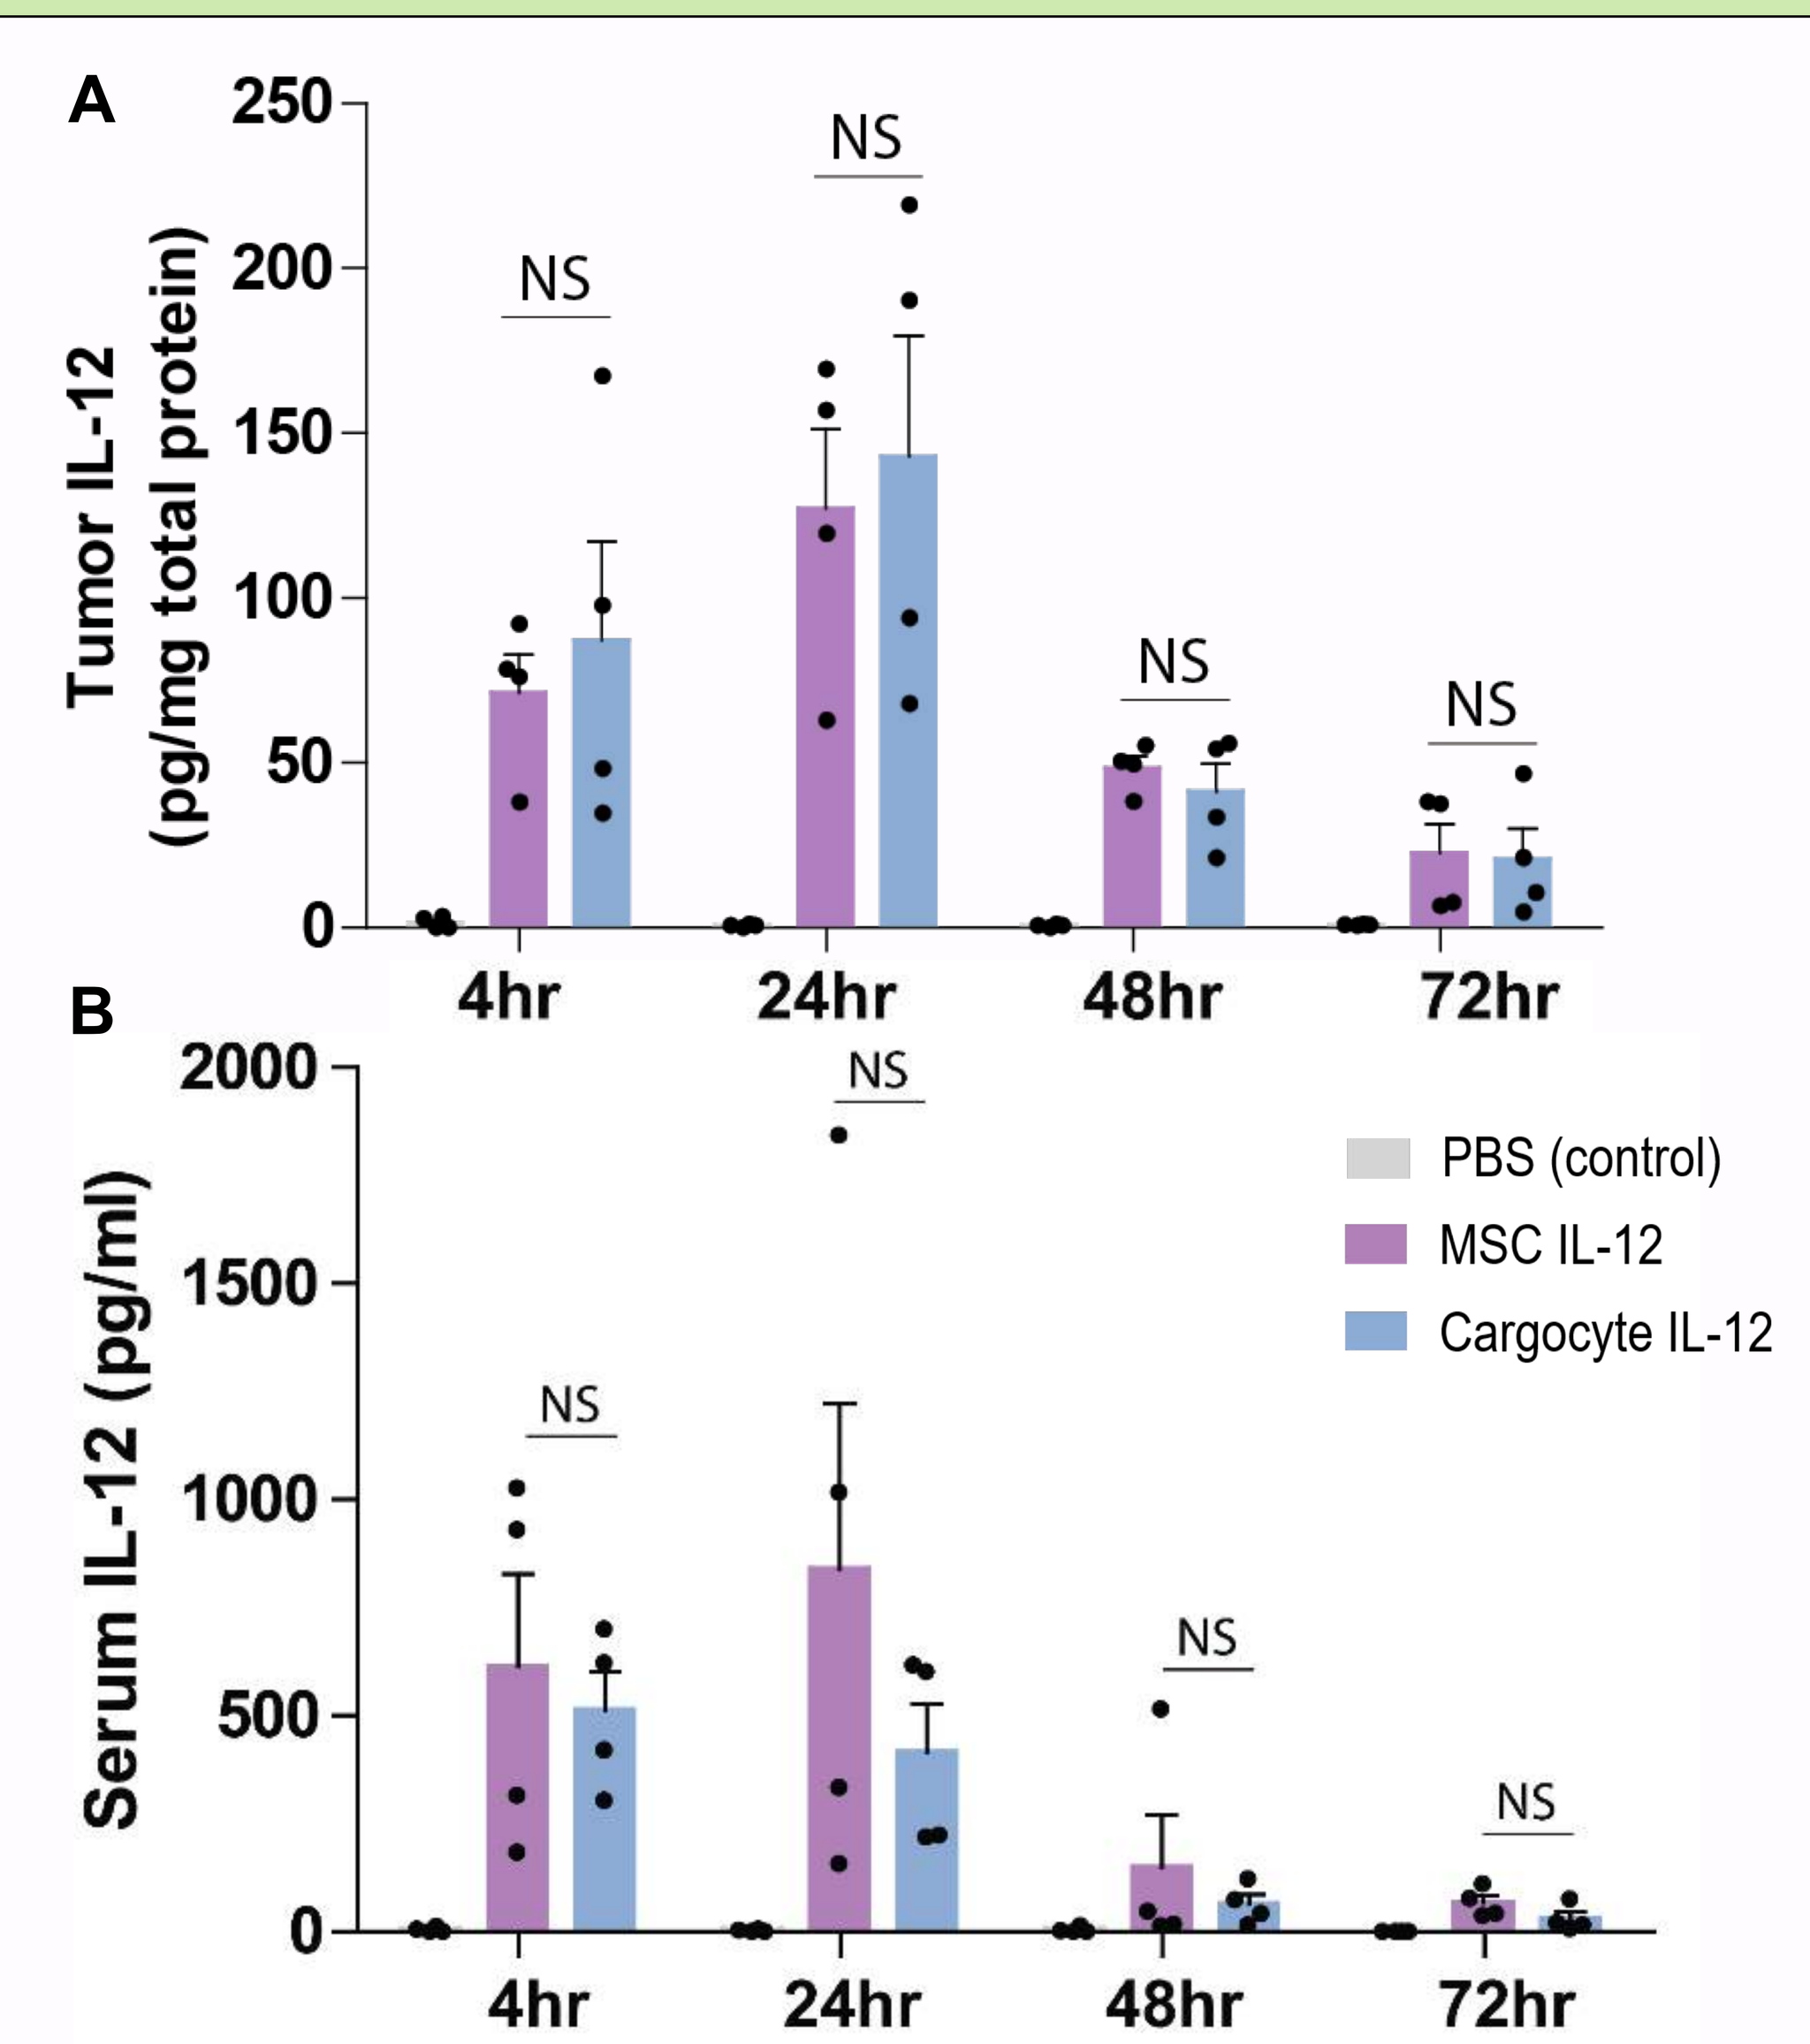
A
Tumor IL-12 (pg/mg total protein)
250
200
150
100
50
0
NS
4hr
24hr
48hr
72hr
B
Serum IL-12 (pg/ml)
2000
1500
1000
500
0
PBS (control)
MSC IL-12
Cargocyte IL-12

Fig 2. Mice bearing subcutaneous E0771 tumors were intratumorally injected with either PBS, MSCs transfected with IL-12 mRNA, or cargocytes transfected with IL-12 mRNA. At indicated timepoints after injection, the IL-12 level was detected by ELISA in the A) tumor (normalized to protein in tumor (pg/mg)) and B) serum (pg/ml). NS =not significant.

Figure 3. Intratumoral injections of cargocytes stimulate immunoregulatory cytokine cascades and increase anti-tumor immune cell recruitment/response

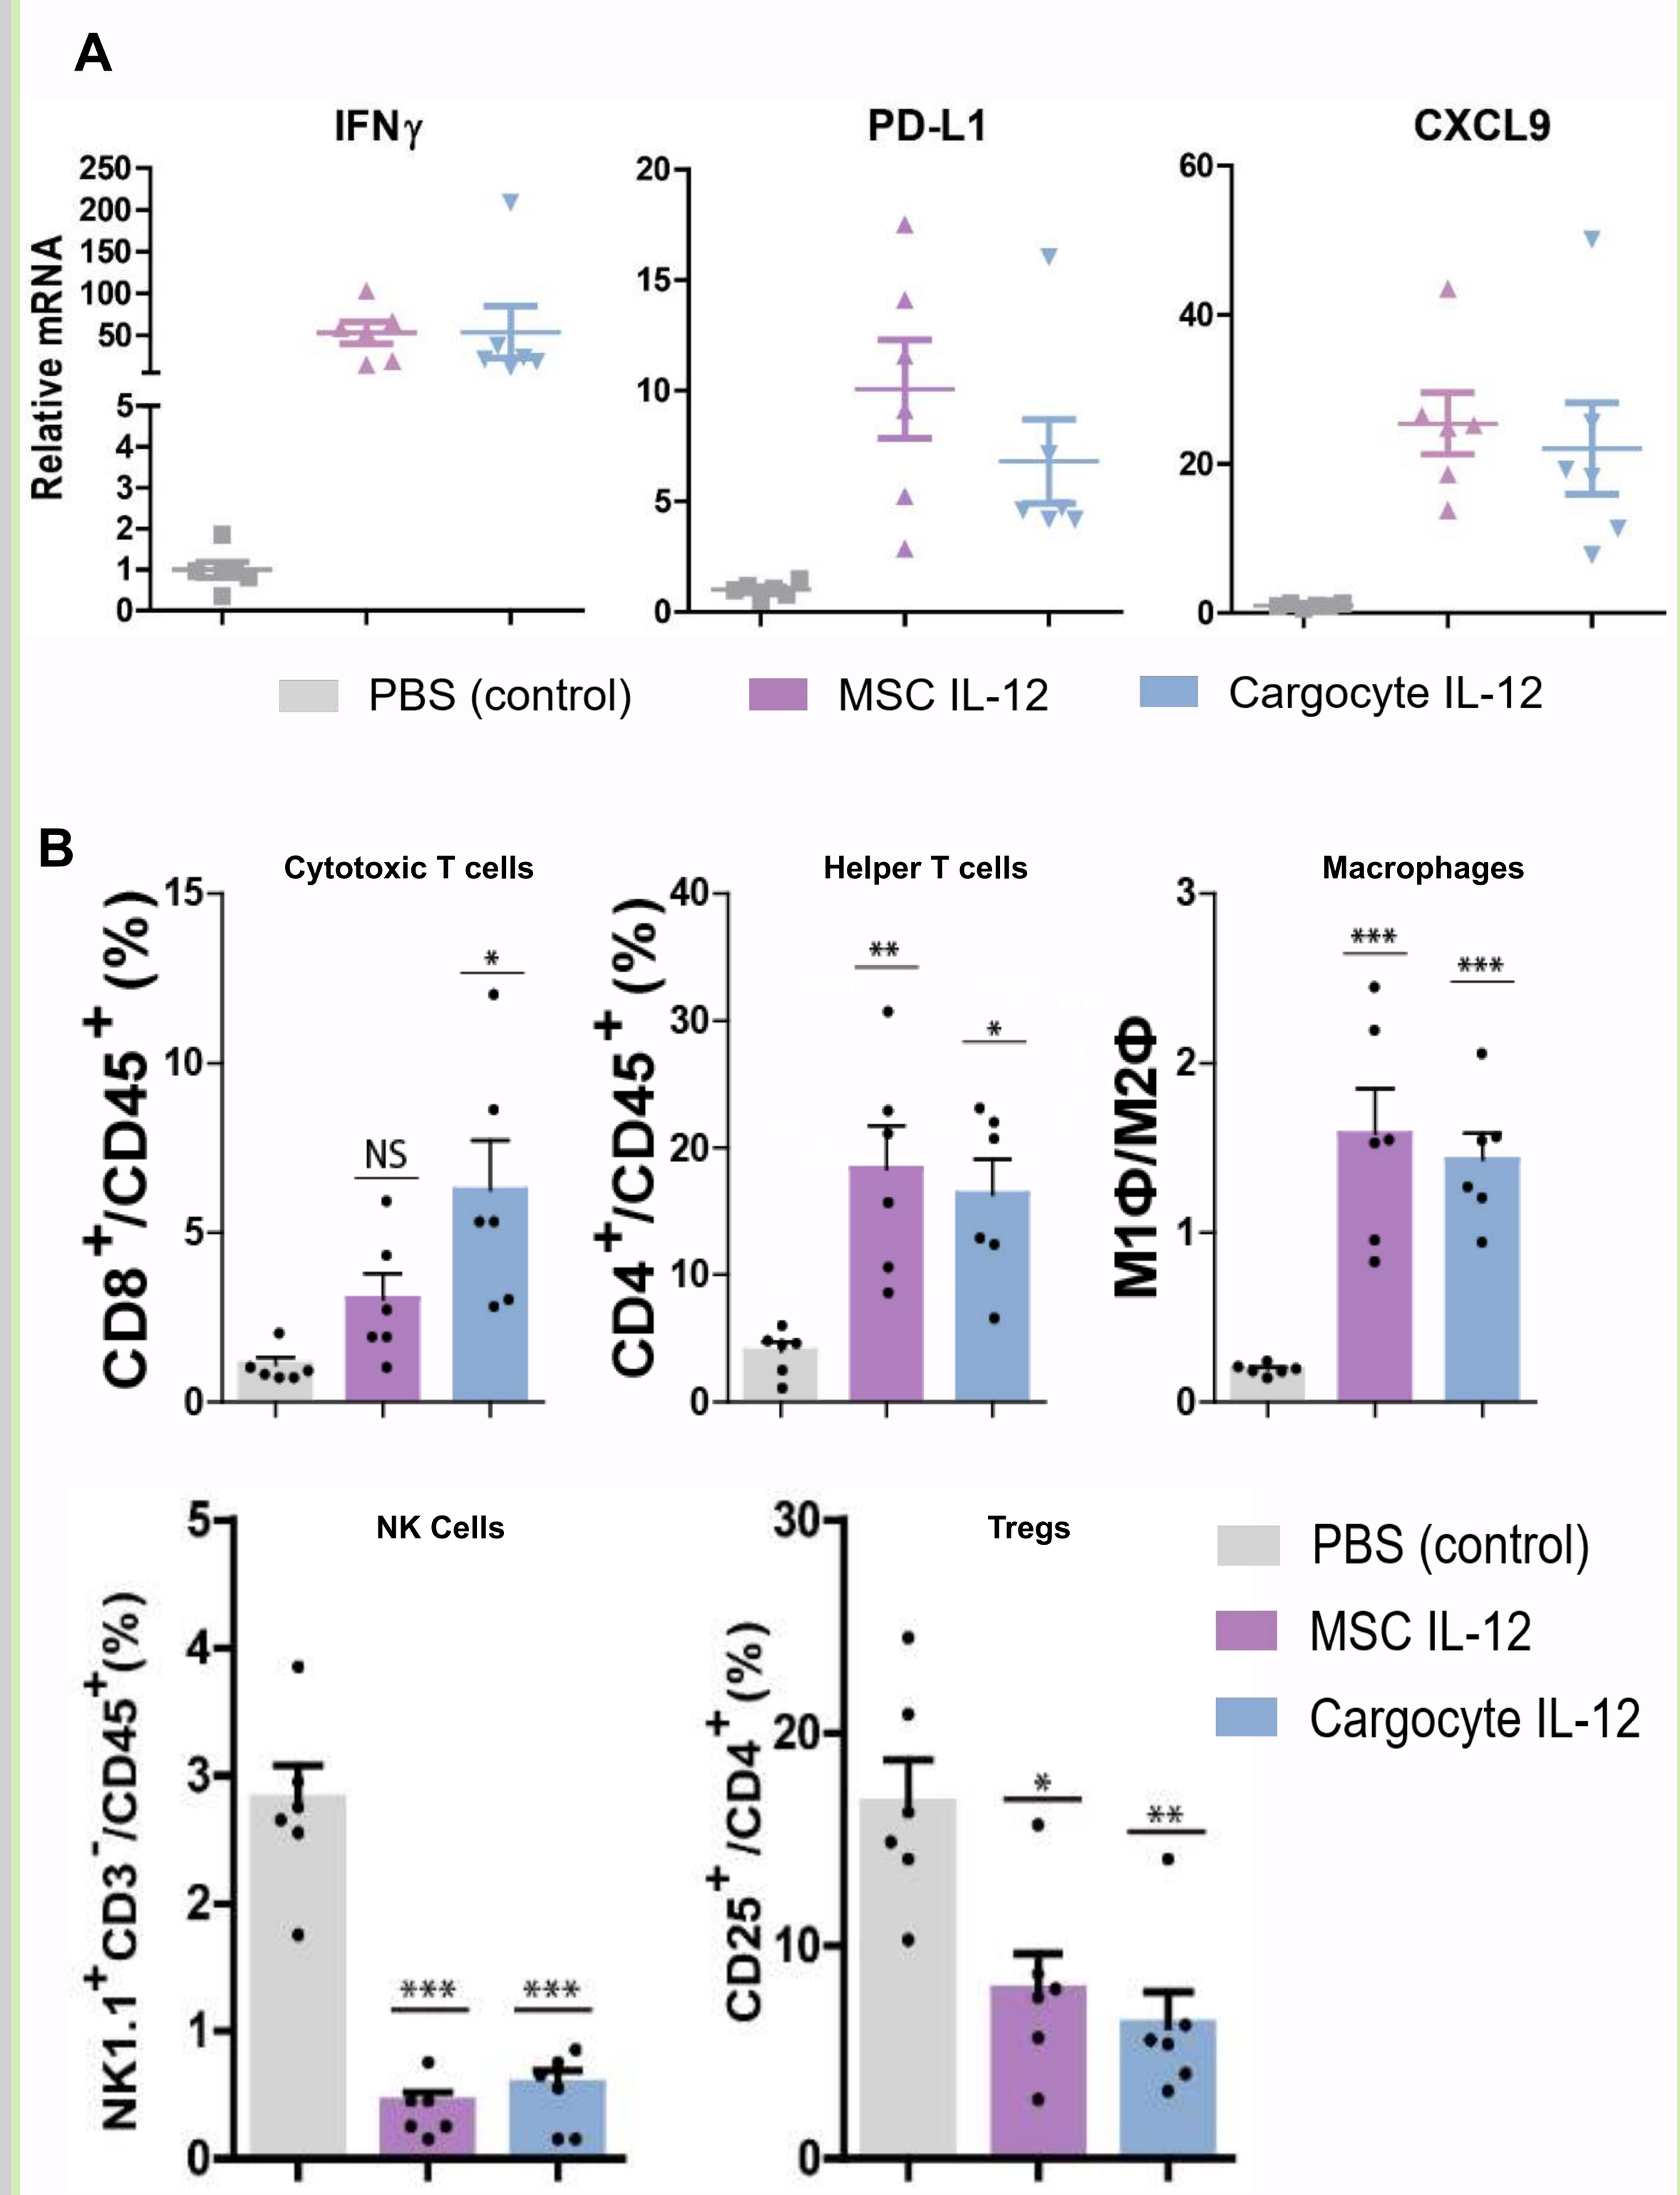
A
IFNγ
PD-L1
CXCL9
Relative mRNA
PBS (control)
MSC IL-12
Cargocyte IL-12
B
Cytotoxic T cells
CD8⁺/CD45⁺ (%)
Helper T cells
CD4⁺/CD45⁺ (%)
Macrophages
M1Φ/M2Φ
NK Cells
NK1.1⁺CD3⁻/CD45⁺(%)
Tregs
CD25⁺/CD4⁺(%)
NS
PBS (control)
MSC IL-12
Cargocyte IL-12

Fig 3. C57Bl/6 mice were injected subcutaneously with 1M E0771 syngeneic mouse breast cancer cells. After ~14 days, established tumors were injected with either 100µl PBS (control), 1M IL-12 transfected MSCs or 3M IL-12 transfected MSC-derived Cargocytes. For experiment A), tumors were harvested 48 after injection, lysed, and analyzed by real-time RT-PCR. Bar graph shows relative fold-change for expression of indicated mRNAs compared to housekeeping HPRT1 between controls, IL-12 transfected MSCs, or IL-12 transfected Cargocytes. For experiment B), mice were intratumorally injected every 2-3 days for a total of 3 injections. 48 hours after the last injection, tumors were analyzed by FACS for the presence of the indicated immune cells.

Figure 4. Cargocyte and checkpoint inhibitor combination therapy design

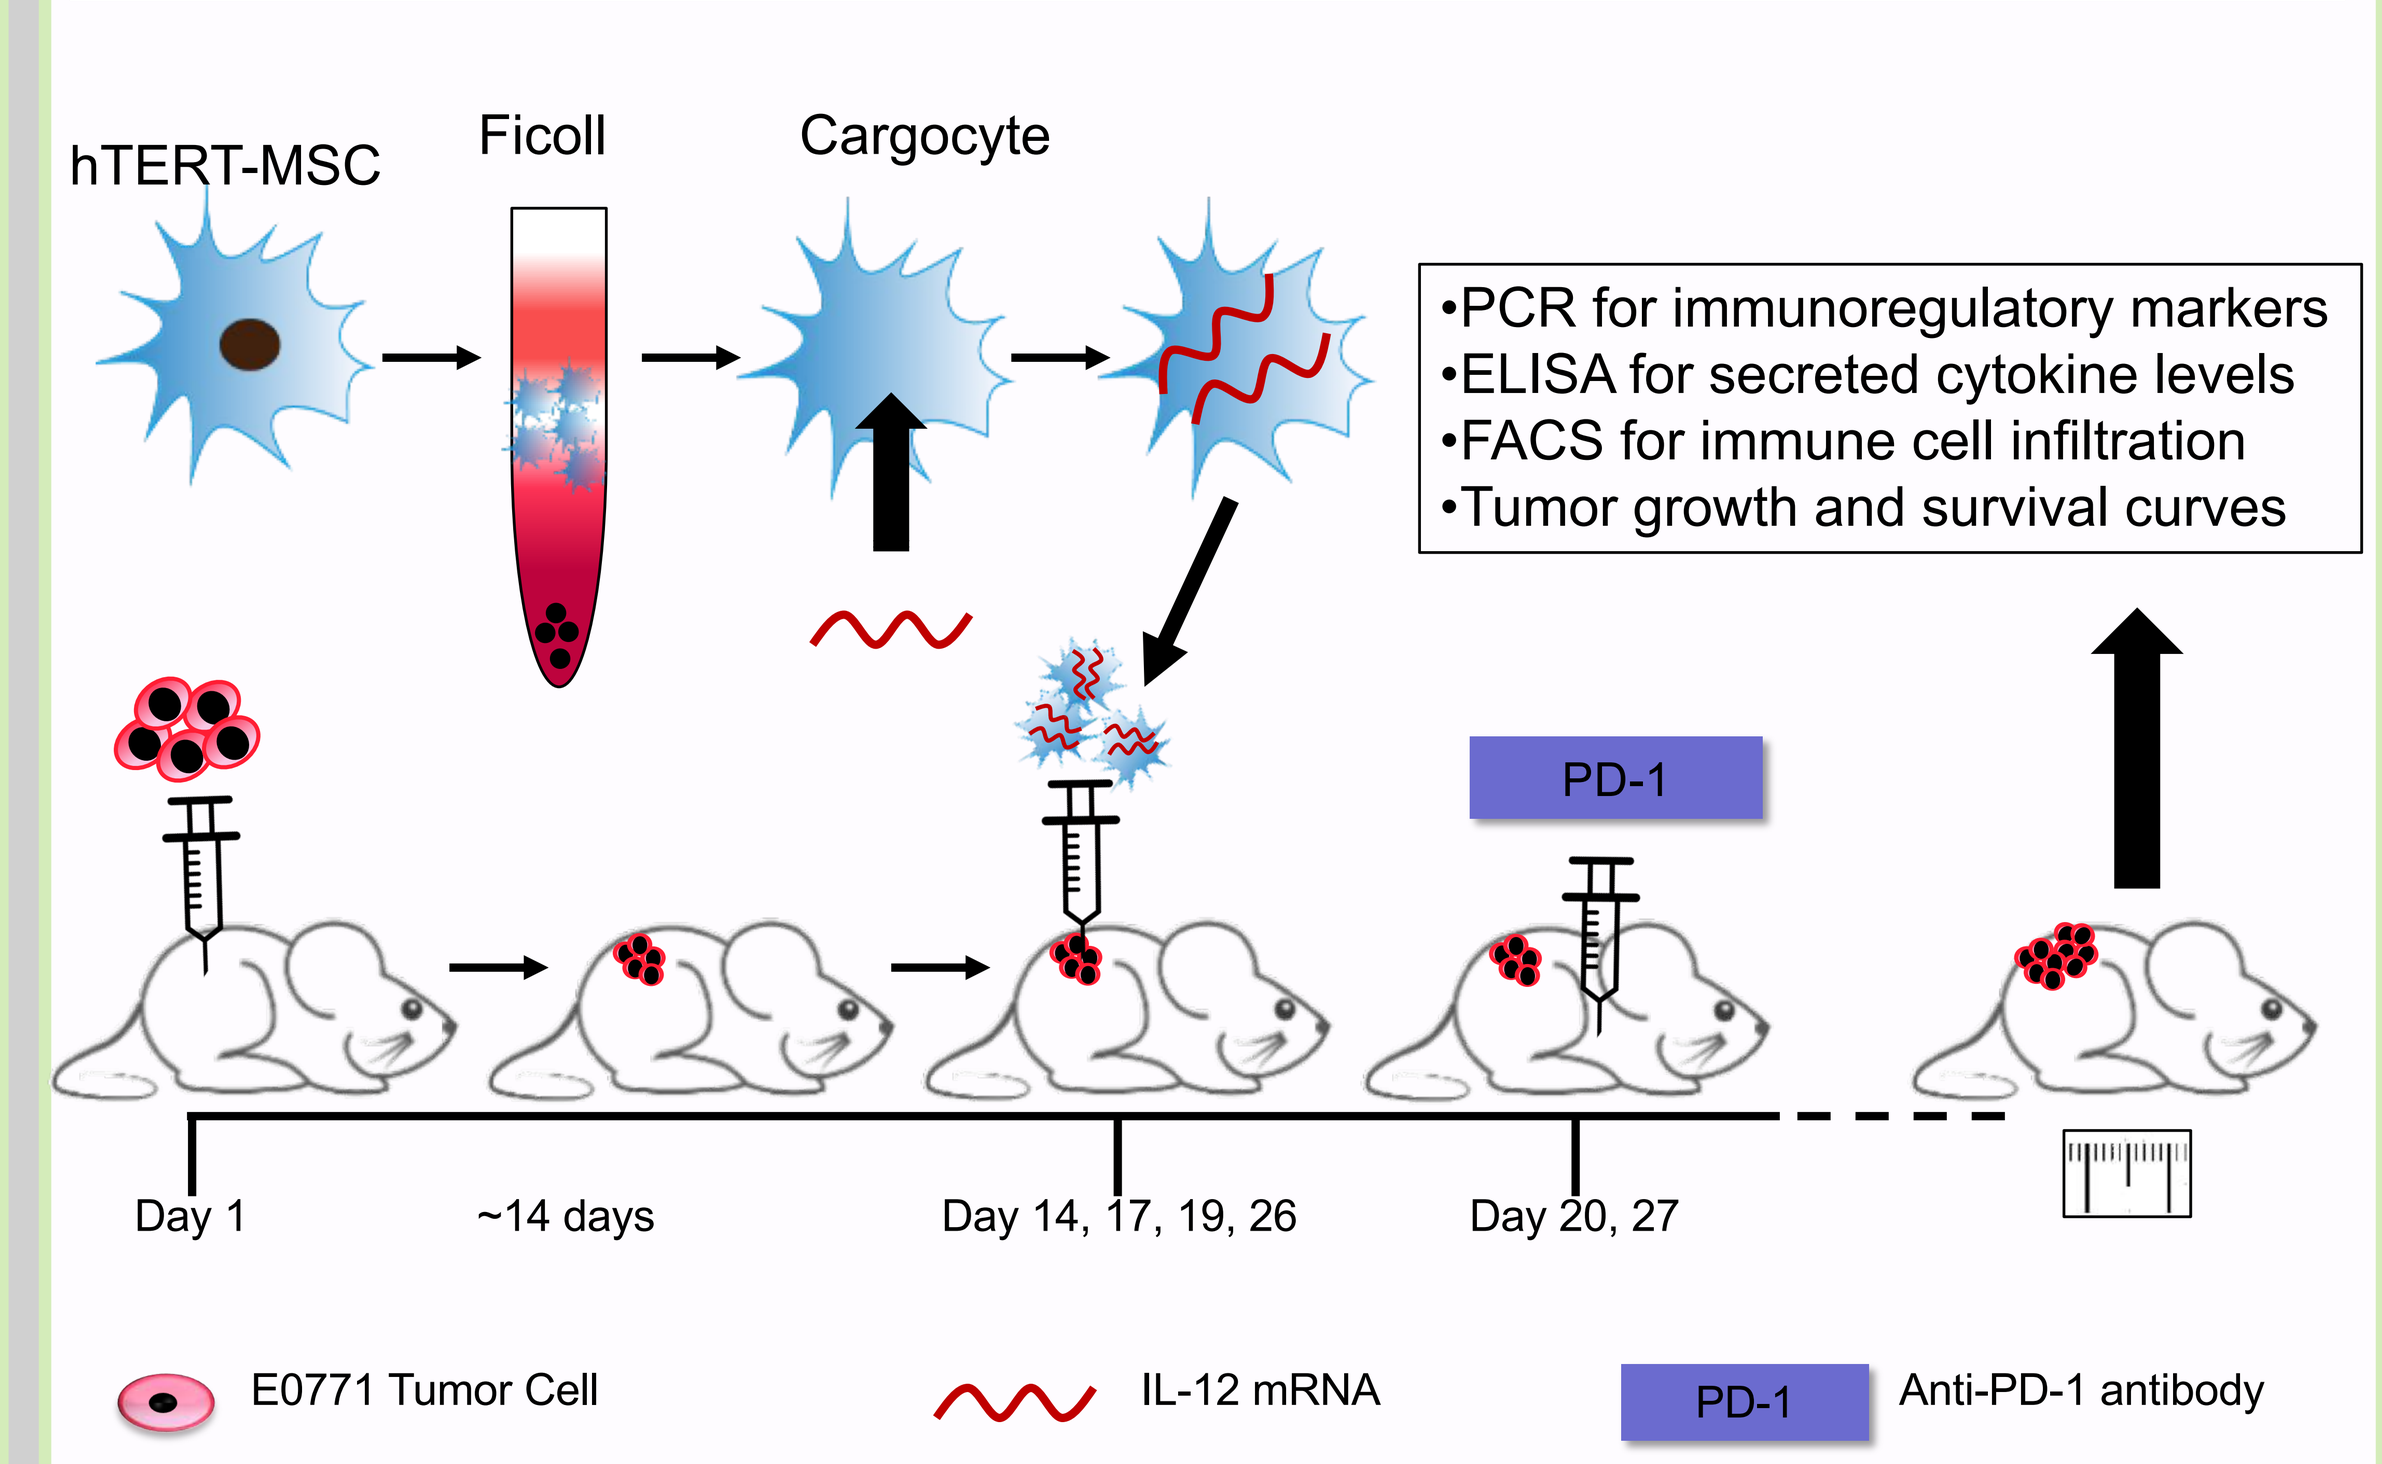
hTERT-MSC
Ficoll
Cargocyte
•PCR for immunoregulatory markers
•ELISA for secreted cytokine levels
•FACS for immune cell infiltration
•Tumor growth and survival curves
PD-1
Day 1
~14 days
Day 14, 17, 19, 26
Day 20, 27
E0771 Tumor Cell
IL-12 mRNA
Anti-PD-1 antibody

Fig 4. Schematic of cargocyte experimental design. C57Bl/6 mice were injected subcutaneously with 1M E0771 syngeneic mouse breast cancer cells. After ~14 days, established tumors were injected with either 100µl PBS (control), 1M IL-12 transfected MSCs or 3M IL-12 transfected MSC-derived cargocytes. Anti-PD-1 antibody or anti-IgG was administered intraperitoneally 24 hours after the third injection. One week later, a fourth injection followed by PD-1/IgG was administered. Animals were monitored at least weekly and tissues collected when tumors reached 2cm diameter in any direction or became ulcerated.

Figure 5. Intratumoral injections of cargocytes with PD-1 inhibitors leads to tumor growth suppression and increased animal survival

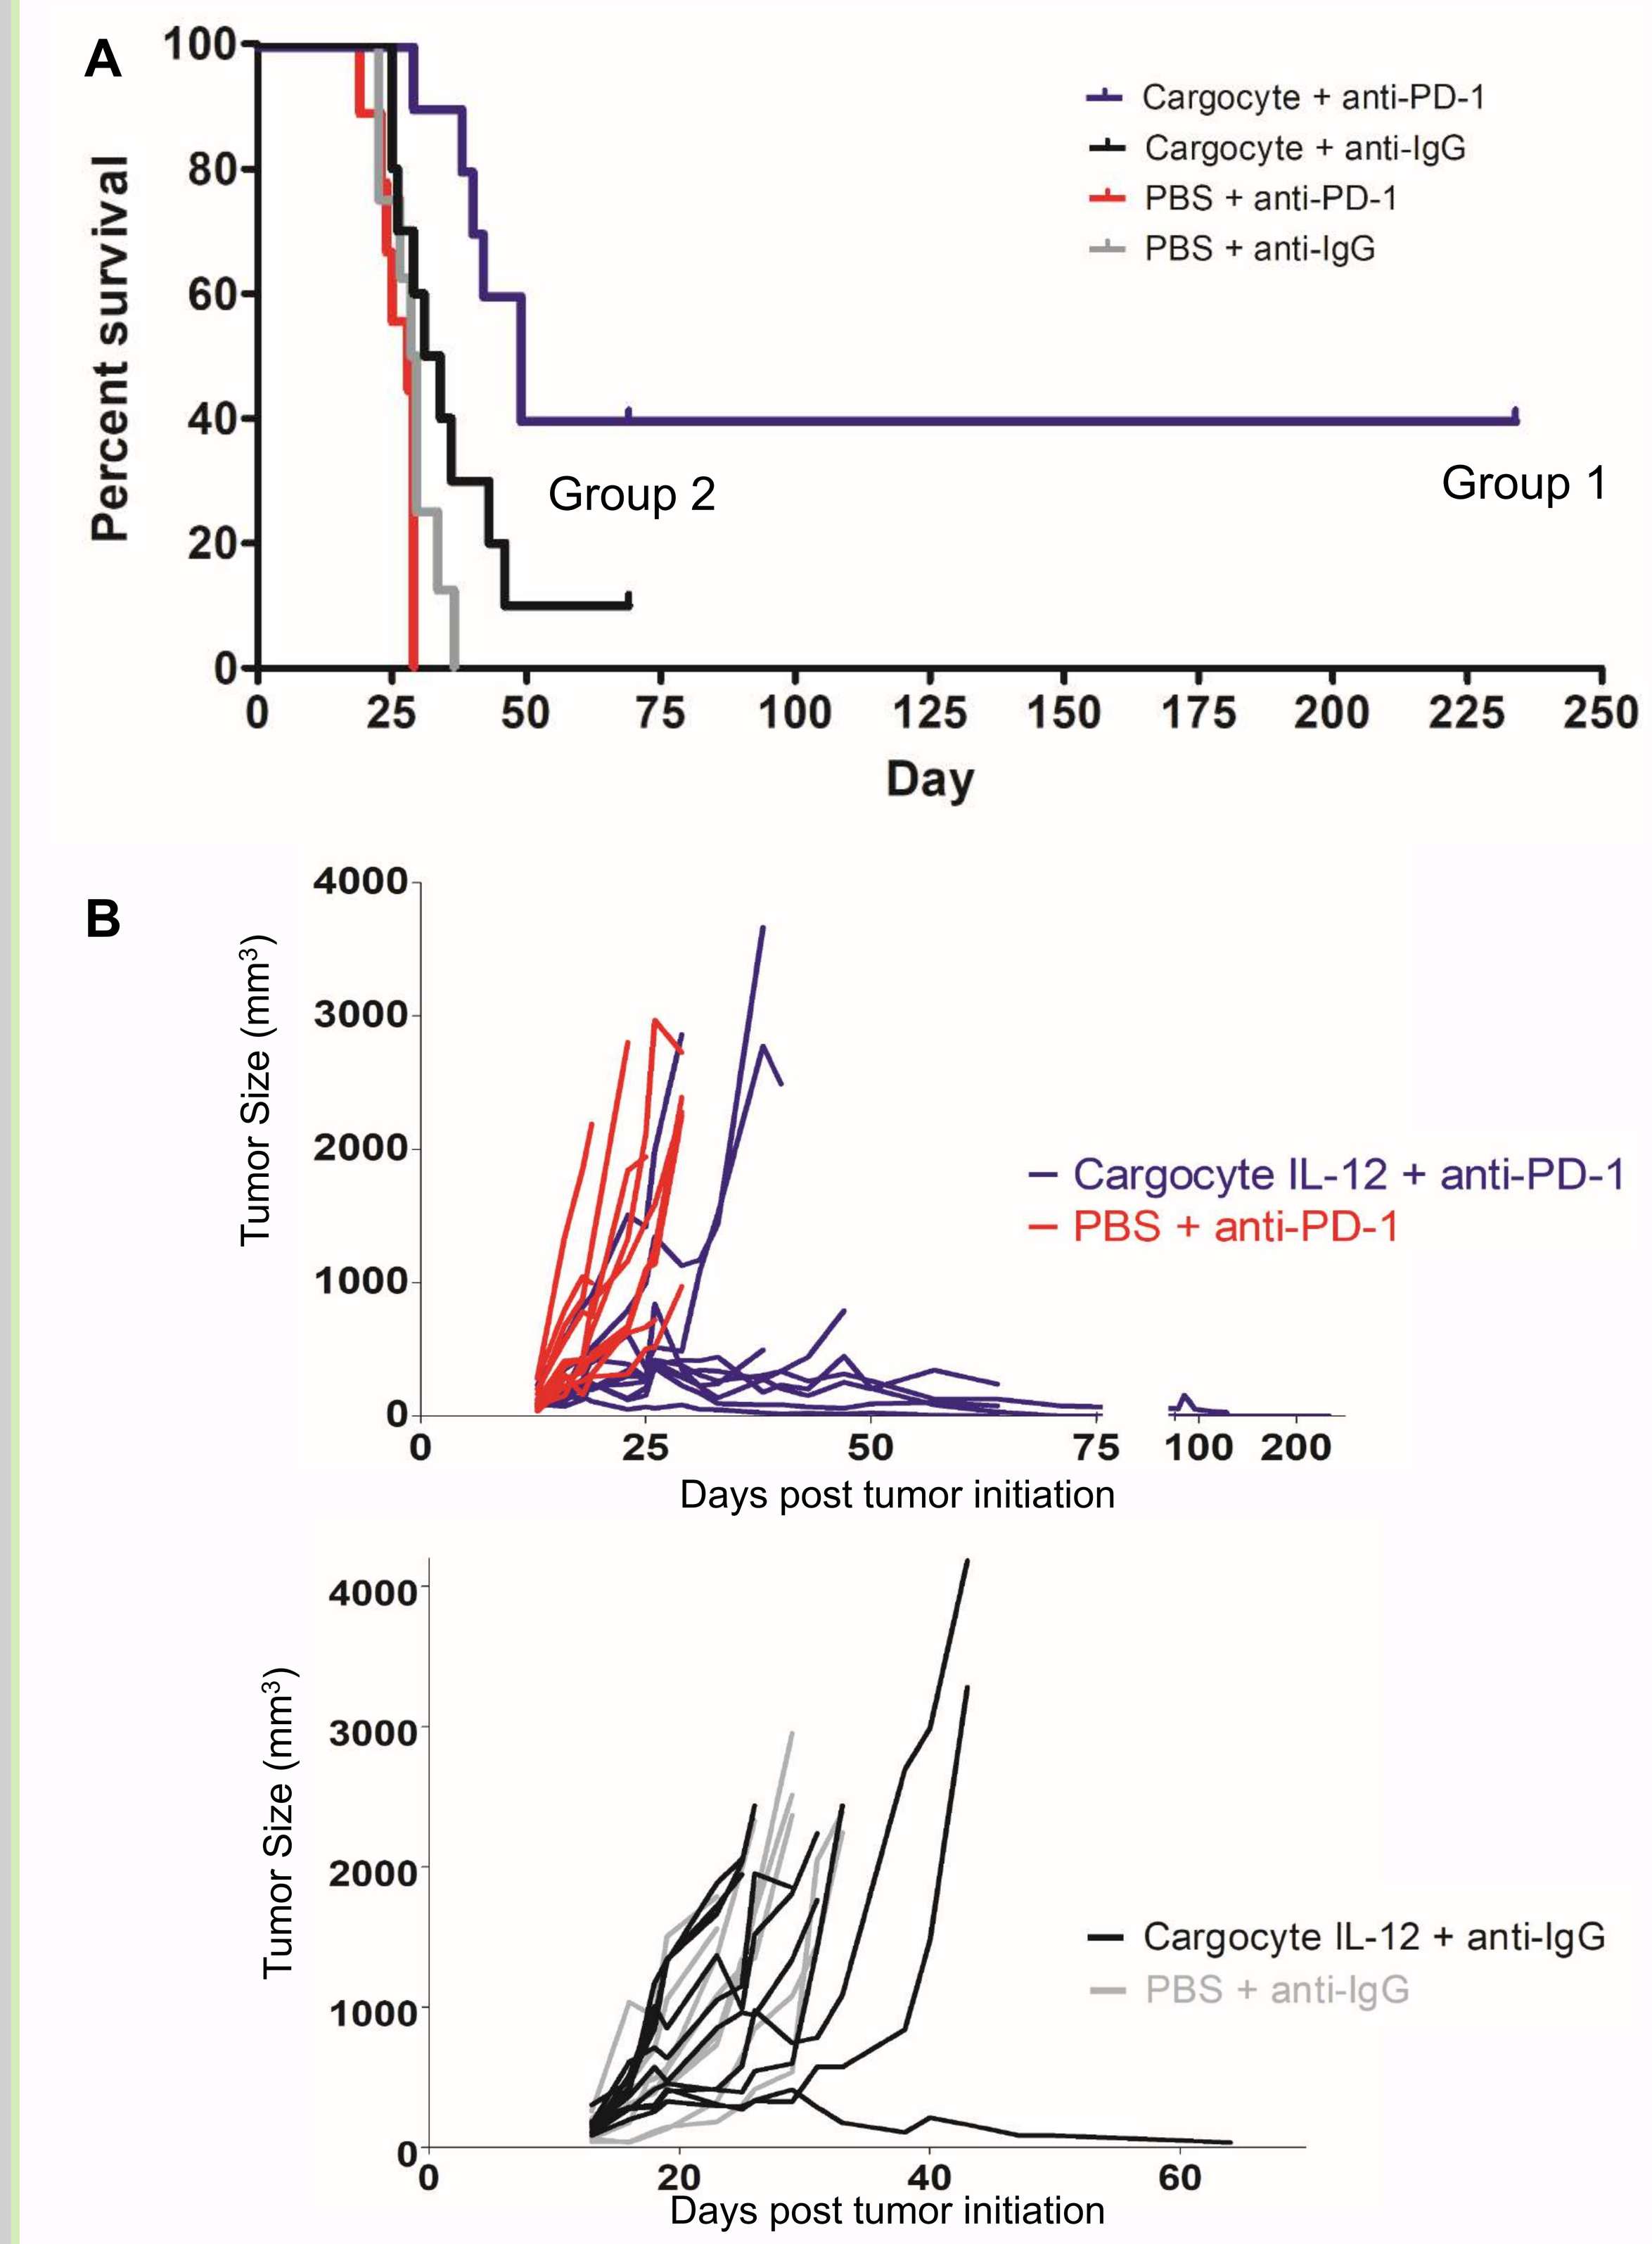
A
Percent survival
Day
Cargocyte + anti-PD-1
Cargocyte + anti-IgG
PBS + anti-PD-1
PBS + anti-IgG
Group 2
Group 1
B
Tumor Size (mm³)
Days post tumor initiation
Cargocyte IL-12 + anti-PD-1
PBS + anti-PD-1
Cargocyte IL-12 + anti-IgG
PBS + anti-IgG

Fig 5. A) Survival curves for animals treated as in Figure 4. N=9-10 (combined from 2 experiments, including both male and female animals. Group 1 animals are currently surviving at 234 days. Group 2 animals are currently surviving at 69 days post tumor initiation. B) Tumor growth curves for animals in 5A by treatment type (anti-PD-1 or anti-IgG).

Conclusions

➢*Cargocytes are a viable cell-like entity for delivery of therapeutic cargo to tumors in vivo.*
- *Secrete IL-12 in the tumor microenvironment*
- *Minimal IL-12 secretion in blood*
- *Induce immunoregulatory cytokines/markers*
- *Recruit anti-tumor immune cells into the tumor*

➢*IL-12 secretion by cargocytes in conjunction with anti-PD-1 therapy suppressed tumor growth and prolonged animal survival in a preclinical breast cancer model.*

Future directions:

➢ **Validate survival effects of cargocyte and combination therapies**
➢ **Investigate long-term tumor immunity in mice with regressed tumors by re-challenge**
➢ **Analyze effects of injection of cargocytes derived from MSCs engineered to express 3 cytokines and multiple anti-tumor antibodies**
➢ **Determine cargocyte homing into subcutaneous and metastatic tumors following intravenous injection**

References

1. Galipeau J, Sensebe J. Mesenchymal Stromal Cells: Clinical Challenges and Therapeutic Opportunities. Cell Stem Cell (2018) 22:824-33.
2. Chen YS, Chen YA, Tsai PH, Chen CP , Shaw SW, Hsuan Y. Mesenchymal Stem Cell: Considerations for Manufacturing and Clinical Trials on Cell Therapy Product. Int J Stem Cell Res Ther (2016) 3:29.
3. Wigler MH, Weinstein IB. A preparative method for obtaining enucleated mammalian cells. BiochemBiophResCo (1975) 63.3:669-74.
4. Colombo MP, Trinchieri G. Interleukin-12 in anti-tumor immunity and immunotherapy. Cytokine Growth Factor (2002) 13: 155–168.
5. Waldmann TA. Cytokines in Cancer Immunotherapy. Cold Spring Harb Perspect Biol (2017) doi: 10.1101/cshperspect.a028472.
6. Johnstone CN, Smith YE, Cao Y, Burrows AD, Cross RS, Ling X, Redvers RP, Doherty JP, Eckhardt BL, Natoli AL, Restall CM, Lucas E, Pearson HB, Deb S, Britt KL, Rizzitelli A, Li J, Harmey JH, Pouliot N, Anderson RL. Functional and molecular characterisation of EO771.LMB tumours, a new C57BL/6-mouse-derived model of spontaneously metastatic mammary cancer. Dis Model Mech. (2015) 8(3):237-51.

Acknowledgements: Work is supported by R.L.K. NIH Grants R01 CA097022, CA184594 and CA182495; C.N.A. was supported by NIH 5T32 OD17863-4. Thank you to Drs. Weian Zhao and Aude Ségaliny (UC Irvine) for discussion.
Conflict of Interest: R.L.K. is a founder and CSO of Cytonus Therapeutics, Inc.