Cargocyte Biofactories: A Novel Platform for Delivering Oncolytic Viruses to Treat Metastatic Cancer

Huawei Wang[1], Bei Liu[1], Christina N. Alarcón[2], Felicia Watson[1], Richard Klemke[1+]
[1]Department of Pathology, University of California, San Diego, La Jolla, CA,[2]Biomedical Sciences Program,, University of California, San Diego, La Jolla, CA
HW, presenter; +Corresponding Author: Richard L. Klemke Ph.D., Department of Pathology, School of Medicine, UCSD 9500 Gilman Drive
#0612, La Jolla, CA 92093. Phone: 858-822-5610; Fax: 858-822-4566; E-mail: rklemke@ucsd.edu

Abstract

Metastatic disease remains the most significant life-threatening event for cancer patients. While intravenous administration of oncolytic viruses (OVs) is a promising method to target tumor cells and metastatic cancers, the immune system can rapidly neutralize intravascular circulating virus hindering overall efficacy towards disseminated tumor cells. Therefore, there is a critical need to design new viral carrier systems that can both specifically deliver OVs to metastatic sites and prevent immune recognition until OVs reach the target site. Mesenchymal stromal cells (MSCs) are attractive delivery vehicles because they have innate tumor trophic (homing) properties and can shield the virus from immune recognition. However, MSC homing efficiency needs to be improved for clinical use, and research has shown that MSCs can engraft in tumors and contribute to tumor growth and metastasis. While genetic and bioengineering strategies can overcome many of these limitations, modification of genomic DNA or introduction of new genetic material into therapeutic cells raises substantial safety concerns, ultimately making FDA approval difficult. To address these issues, we developed a novel, safe, cell-based platform whereby genetically engineered MSCs-derived vehicles can home to metastatic tumors and deliver OVs. We genetically engineered hTERT-immortalized MSCs (hTERT-MSCs) to express chemokine receptors CXCR4 and CCR2, which improve cell homing towards gradients of SDF-1α and CCL2 respectively. These chemokines are commonly increased in inflamed stroma and produced by tumor cells. Additionally, the MSCs were engineered to co-express PSGL-1 and α1,3/4-fucosyltransferase to mediate hTERT-MSC adhesion to inflamed endothelial cells in the tumor vasculature. The genetically modified nuclei of these engineered cells were subsequently removed using an optimized enucleation protocol and Ficoll density centrifugation. These enucleated cells (Cargocytes™) have greatly improved safety profiles compared to nucleated MSCs because they do not permanently engraft into the body nor do they transfer any dangerous genomic DNA to the patients. Cargocytes retain many desired biological functions (endogenous and engineered), such as viability up to 72 hours, retention of cell surface markers/proteins, translation and secretion of bioactive molecules, and robust *in vitro* and *in vivo* homing. Cargocyte therapeutics are highly versatile biofactories that can be engineered to produce or to be exogenously loaded with many important biologics including secreted cytokines, RNA-based therapeutics, peptides, small molecule drugs, and importantly, OVs. When infected with OVs, Cargocytes show dramatically increased OV production due to lack of transcription-dependent host defense mechanisms against viruses. In fact, Cargocytes harboring oncolytic vesicular stomatitis virus (oVSV) robustly propagate, package, and release functional virus capable of infecting and killing tumor cells. In *in vitro* homing assays and in a preclinical mouse model of metastatic breast cancer, Cargocytes harboring oVSV retain chemokine homing potential and their ability to transfer virus to lung metastases. Overall, our findings demonstrate that Cargocyte biofactories are a genetically tractable cell therapy platform that is scalable for clinical use, and has potential to improve systemic delivery of OVs alone or in combination with other biologics for the treatment of metastatic cancer.

Background

➢Triple Negative Breast Cancers (TNBC: ER⁻, PR⁻, HER2/neu⁻) are often highly metastatic with poor prognosis under current therapies[1].

➢Oncolytic viruses (OV) such as oMV (Measles Virus), oHSV (Herpes simplex virus), and oVSV (Vesicular stomatitis virus) hold great promise as anti-cancer treatments, including metastatic TNBC[2,3].

➢Systematic administration of OVs into patients is highly inefficient because the immune system rapidly clears unprotected viruses [4].

➢Therefore, there is a major need to develop biocompatible carriers that improve systemic delivery of oncolytic viruses and target them to primary and metastatic tumors in a safe manner [5,6].

➢ To study the *in vivo* tumor-delivery of OVs, a syngeneic TNBC model in immuno-competent mice is ideal.

➢Our platform technology enucleates hTERT-immortalized MSCs to generate nucleus-free cargocytes, which can be extensively engineered with improved homing abilities to safely deliver oncolytic viruses to tumors.

Strategy

Cargocytes™ are therapeutic, enucleated cells that retain functional organelles, cell-like properties, and engineered capabilities

A **Generation of Cargocytes: Enucleation procedure**

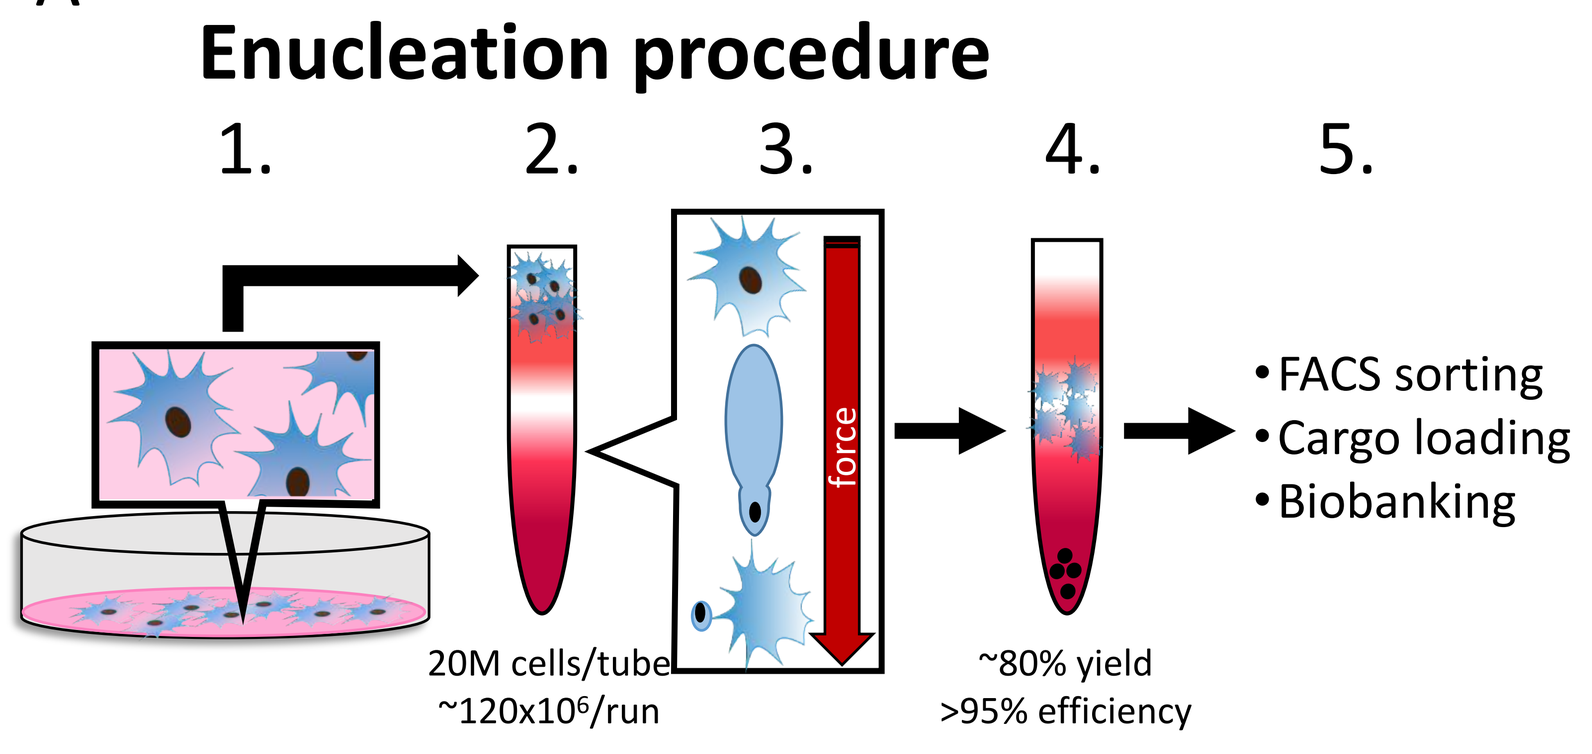
1.
2.
3.
4.
5.
force
•FACS sorting
•Cargo loading
•Biobanking
20M cells/tube
~120x10⁶/run
~80% yield
>95% efficiency

B **Cargocyte Schematic**

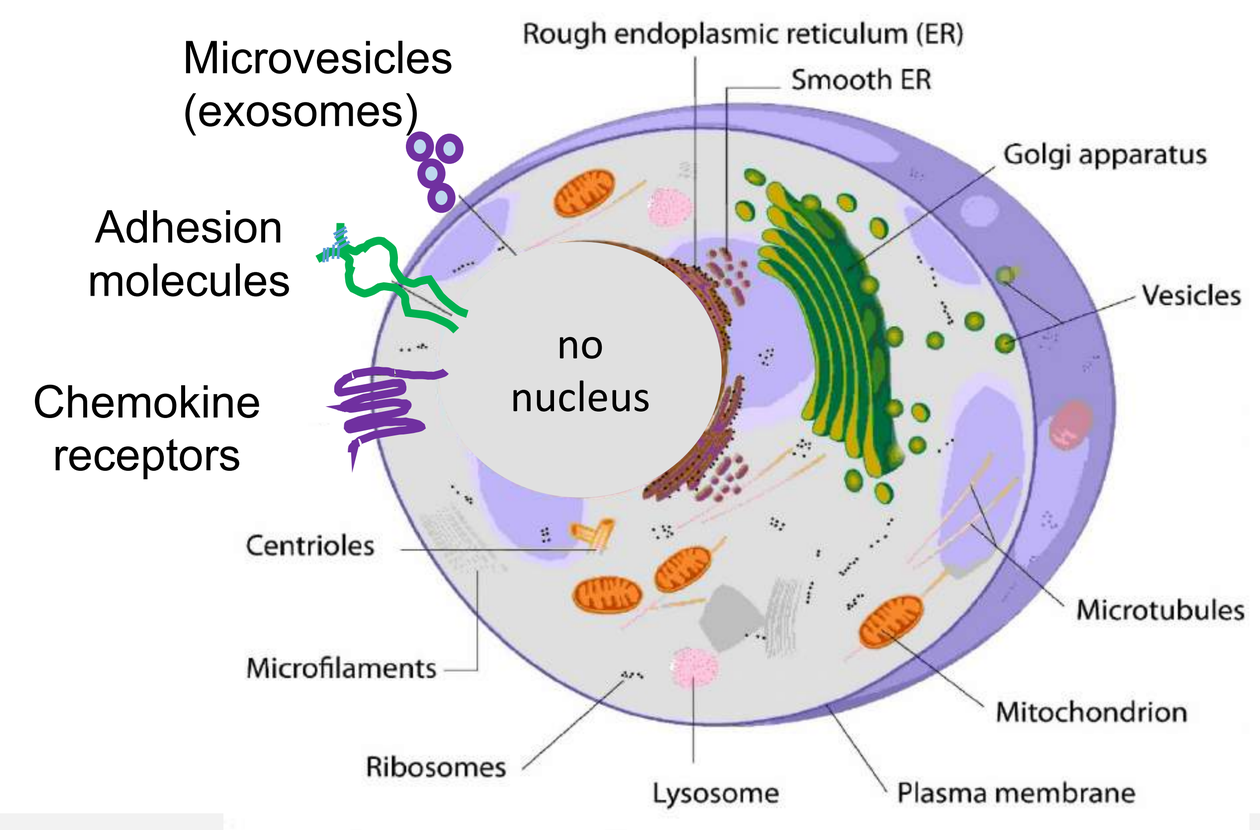
Microvesicles (exosomes)
Rough endoplasmic reticulum (ER)
Smooth ER
Golgi apparatus
Adhesion molecules
Vesicles
no nucleus
Chemokine receptors
Centrioles
Microtubules
Microfilaments
Mitochondrion
Ribosomes
Lysosome
Plasma membrane

Enucleation procedure to generate Cargocytes:
1. Cell expansion in culture
2. Cells suspended in Ficoll gradients
3. Ultracentrifugation pulls nucleus out of cell
4. Therapeutic enucleated cells (Cargocytes™) harvested from Ficoll layer
5. Cargocyte purification, additional engineering

Key Features of Cargocytes:
- Survive up to 72 hours in culture, and survive cryopreservation and cryohibernation
- Retain functional ER, Golgi, mitochondria, microvesicles, cytoskeleton, and cell surface proteins
- Migrate and home towards chemoattractants in vitro and in vivo (FBS, PDGF-AB, IGF-1, SDF-1a, CCL2)
- Translate endogenous and exogenous mRNAs into functional proteins (Gluc, IL-10, IL-12, GFP, GM-CSF)
- Can be stably engineered to express homing receptors and adhesion molecules (CXCR4, CCR2, PSGL-1) for up to 72 hours
- Can be loaded with various cargoes (chemotherapeutics, membrane internalizing peptides, siRNA, shRNA, mRNAs, oncolytic viruses)
- Produce detectable amounts of cargo in tissue microenvironments (intratumoral IL-12, SQ IL-10)
- Change tissue microenvironments (increase tumor-infiltrating leukocytes, alter production of inflammatory cytokines)
- Smaller size compared to cells means reduced entrapment in the lungs

C **Cargocyte Engineering Workflow**

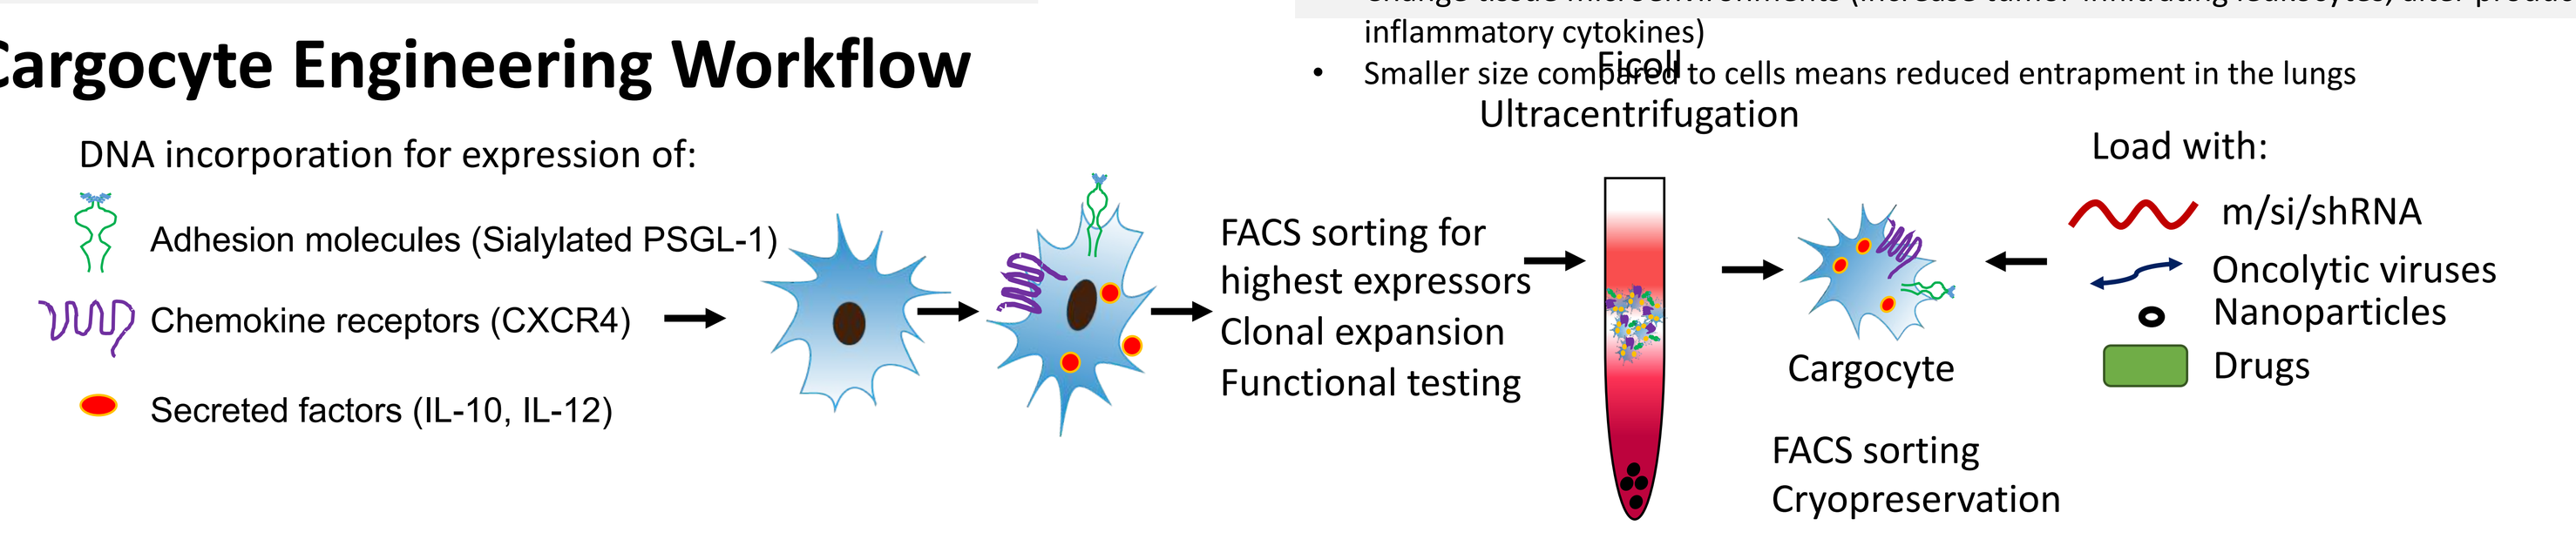
DNA incorporation for expression of:
Adhesion molecules (Sialylated PSGL-1)
Chemokine receptors (CXCR4)
Secreted factors (IL-10, IL-12)
FACS sorting for highest expressors
Clonal expansion
Functional testing
Ficoll
Ultracentrifugation
Cargocyte
FACS sorting
Cryopreservation
Load with:
m/si/shRNA
Oncolytic viruses
Nanoparticles
Drugs

oVSV-ΔM51-GFP propagation in MSC or MSC-derived cargocytes.

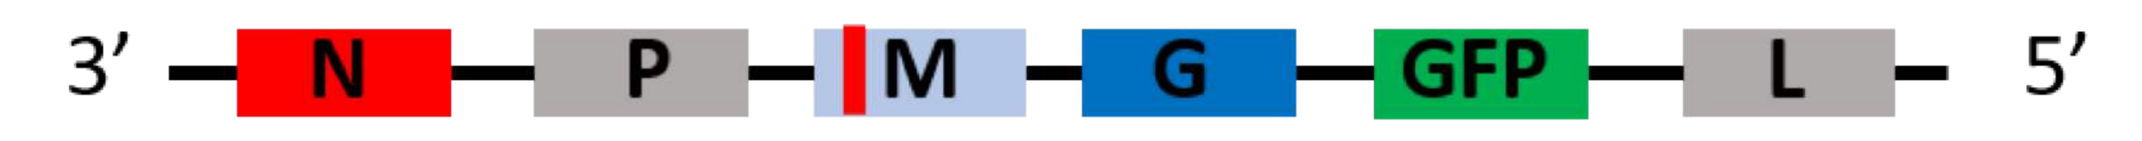
A
3'
N
P
M
G
GFP
L
5'

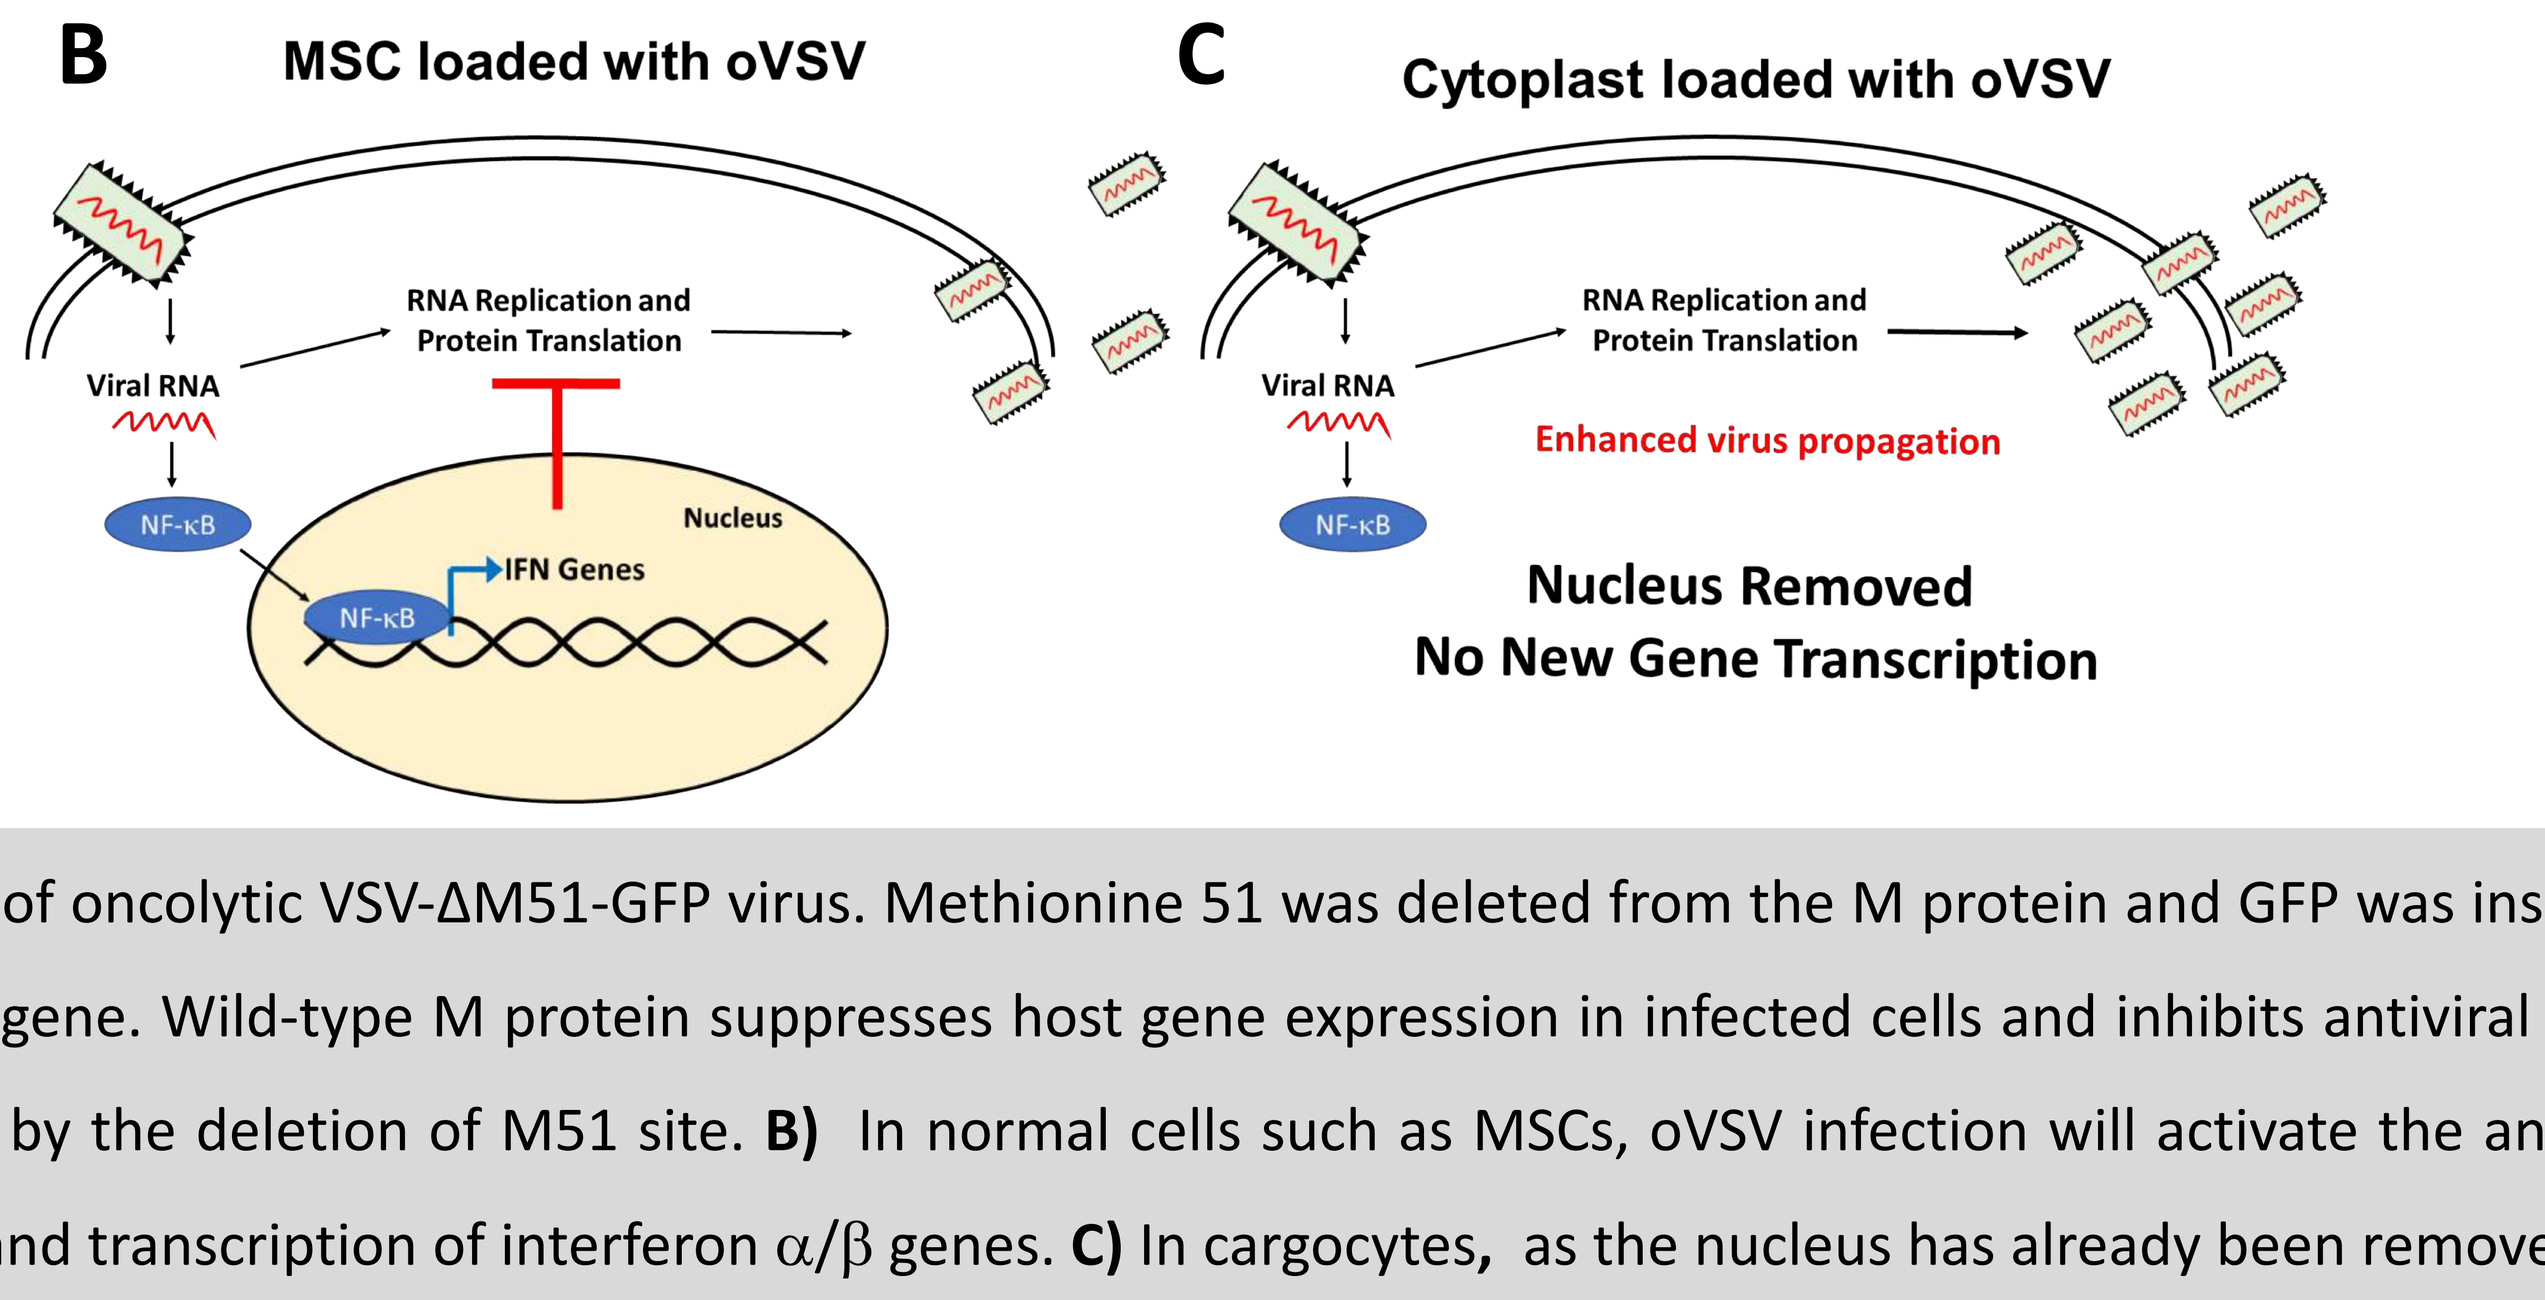
B
MSC loaded with oVSV
Viral RNA
RNA Replication and Protein Translation
NF-κB
Nucleus
IFN Genes
C
Cytoplast loaded with oVSV
Enhanced virus propagation
Nucleus Removed
No New Gene Transcription

A) Schematic design of oncolytic VSV-ΔM51-GFP virus. Methionine 51 was deleted from the M protein and GFP was inserted upstream of the L polymerase gene. Wild-type M protein suppresses host gene expression in infected cells and inhibits antiviral responses. This activity is abrogated by the deletion of M51 site. **B)** In normal cells such as MSCs, oVSV infection will activate the antiviral response mediated by NF-κB and transcription of interferon α/β genes. **C)** In cargocytes, as the nucleus has already been removed, there will be no new gene transcription, which leads to enhanced virus propagation.

Figure 1. Cargocytes can be infected with oVSV- ΔM51-GFP at high efficiency and have functional virus propagation.

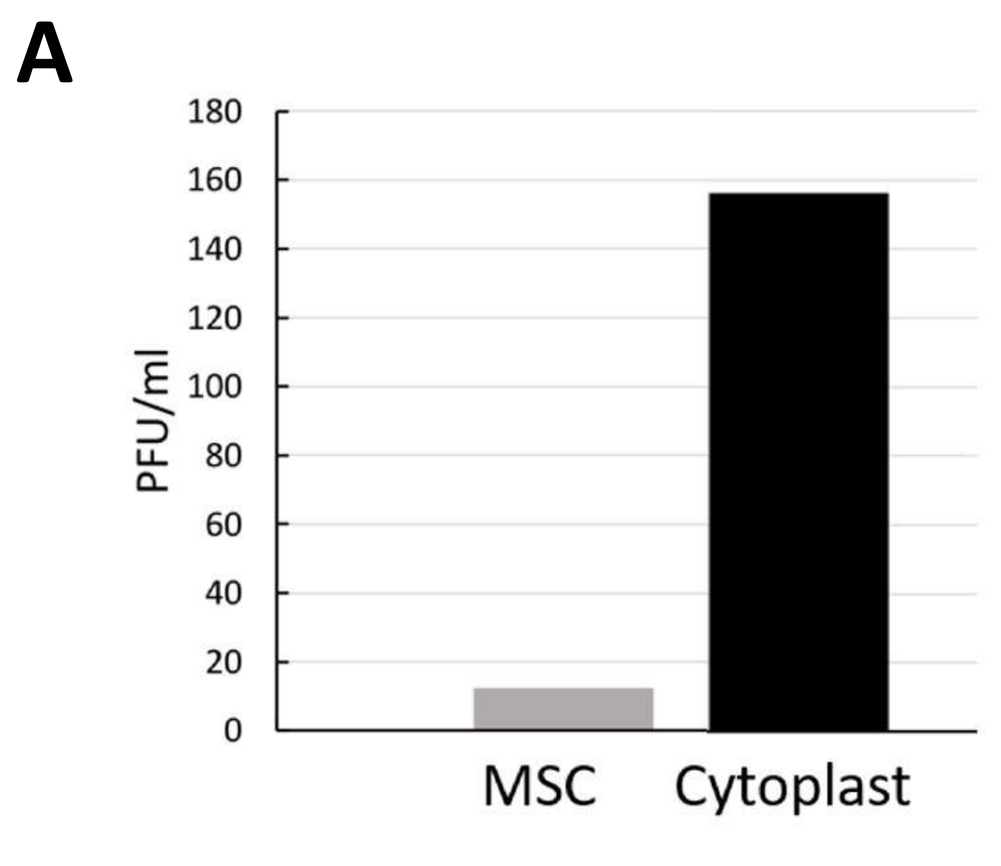
A
180
160
140
120
100
80
60
40
20
0
PFU/ml
MSC
Cytoplast

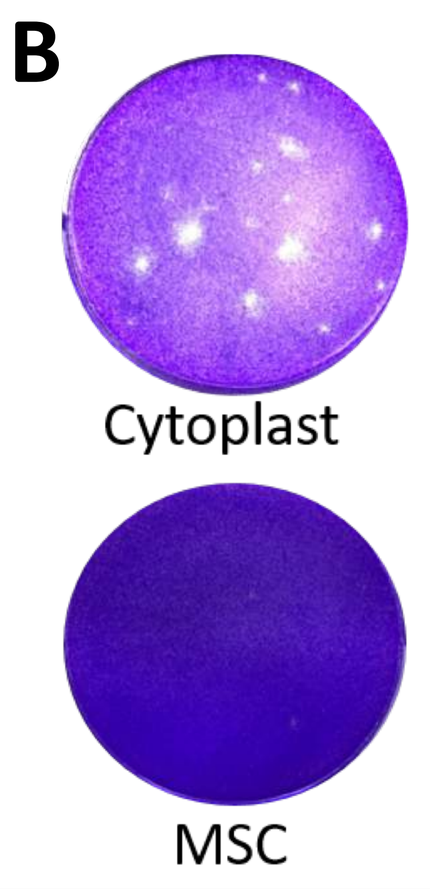
B
Cytoplast
MSC

A) 25,000 MSCs and Cargocytes were infected with oVSV-ΔM51-GFP at MOI 0.1. 72 hours after infection, the plaque forming units (PFUs) from the supernatants of each group were determined by plaque assay. **B)** Representative image of Crystal violet-stained plaque assay wells in which 600,000 Vero cells were seeded into each 6-well. 24 hours later, cells were infected with 10-fold dilution of supernatants from oVSV-ΔM51-GFP-infected cargocytes or MSCs. n=3

Figure 2. oVSV-ΔM51-GFP infects MSC-derived cargocytes and E0771 cancer cells.

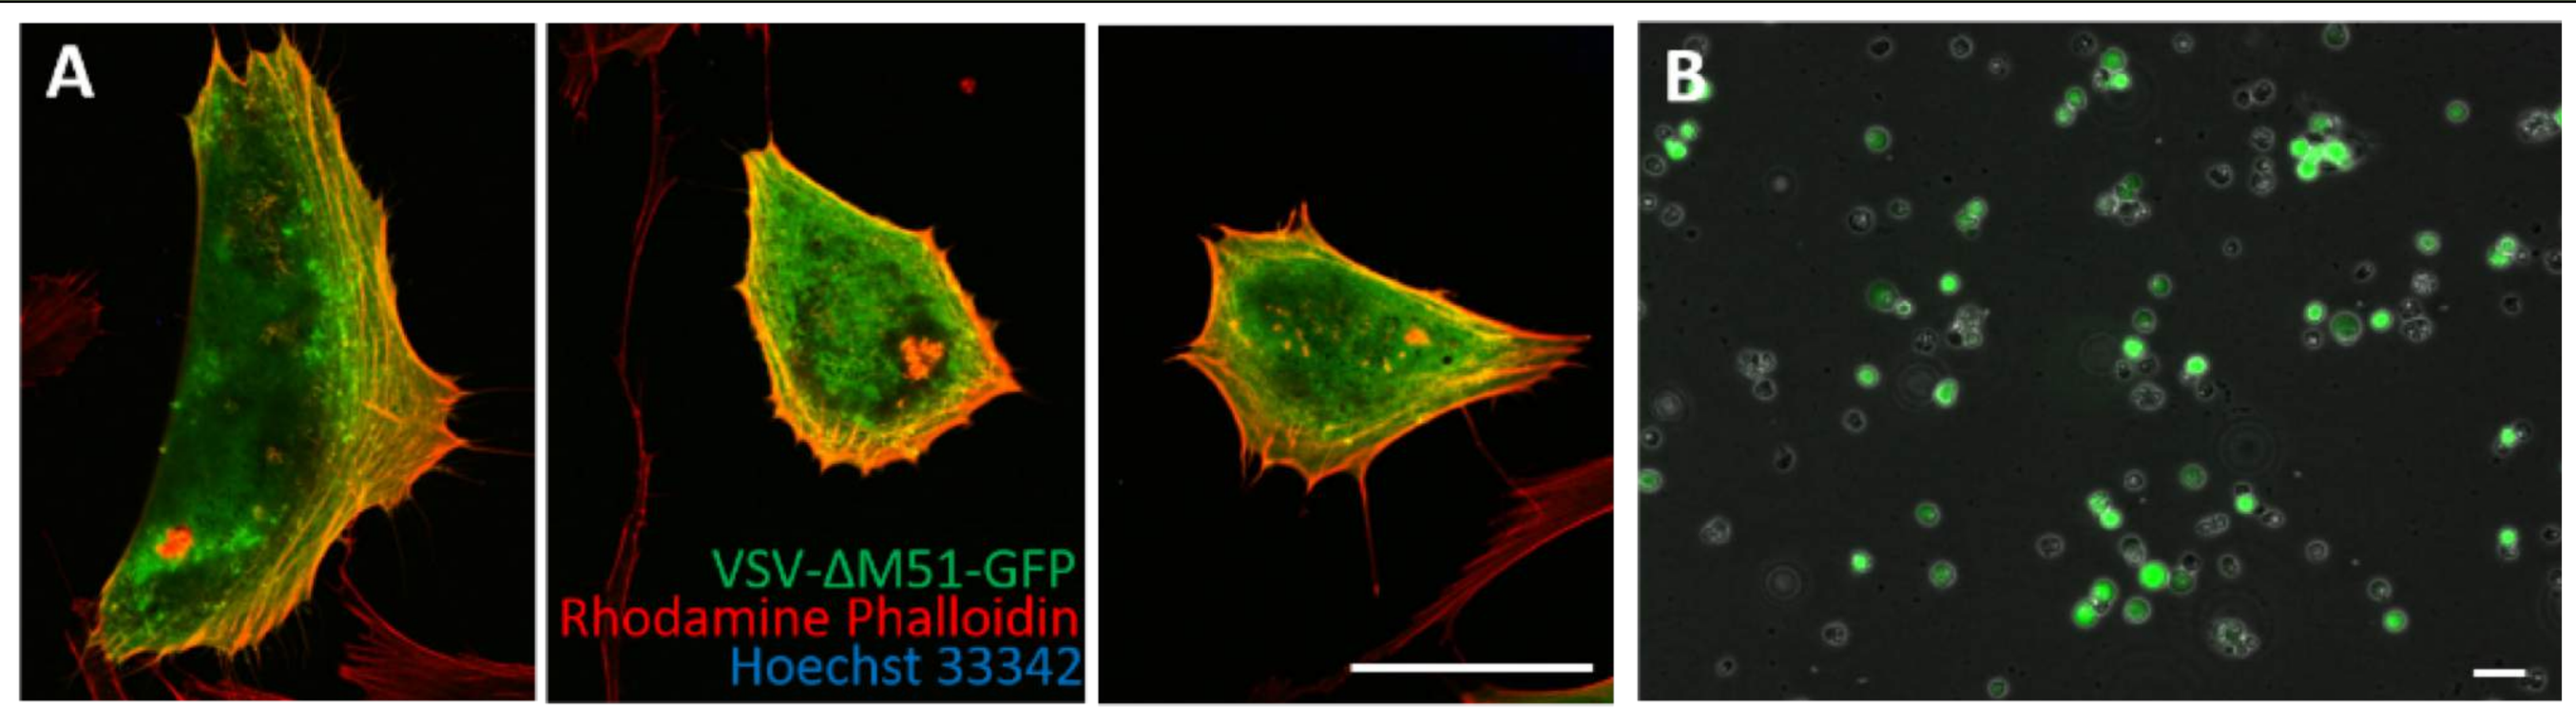
A
B
VSV-ΔM51-GFP
Rhodamine Phalloidin
Hoechst 33342

A) 25,000 cargocyte were infected with oVSV-ΔM51-GFP at MOI 0.1. After 48 hours, cargocytes were fixed, stained with anti-GFP, rhodamine phalloidin and Hoechst 33342 for confocal microscopy. Merged images of 3 representative cargocytes shown. Note the absence of Hoechst (no nuclei). Scale bar = 50 µm. **B)** 100,000 E0771 cells were infected with oVSV-ΔM51-GFP at MOI 0.1. Merged phase contrast and FITC channel images were taken 24 hours after infection. Scale bar = 50 µm.

Figure 3. 3D-cultured MSC-derived cargocytes have improved *in vivo* biodistribution

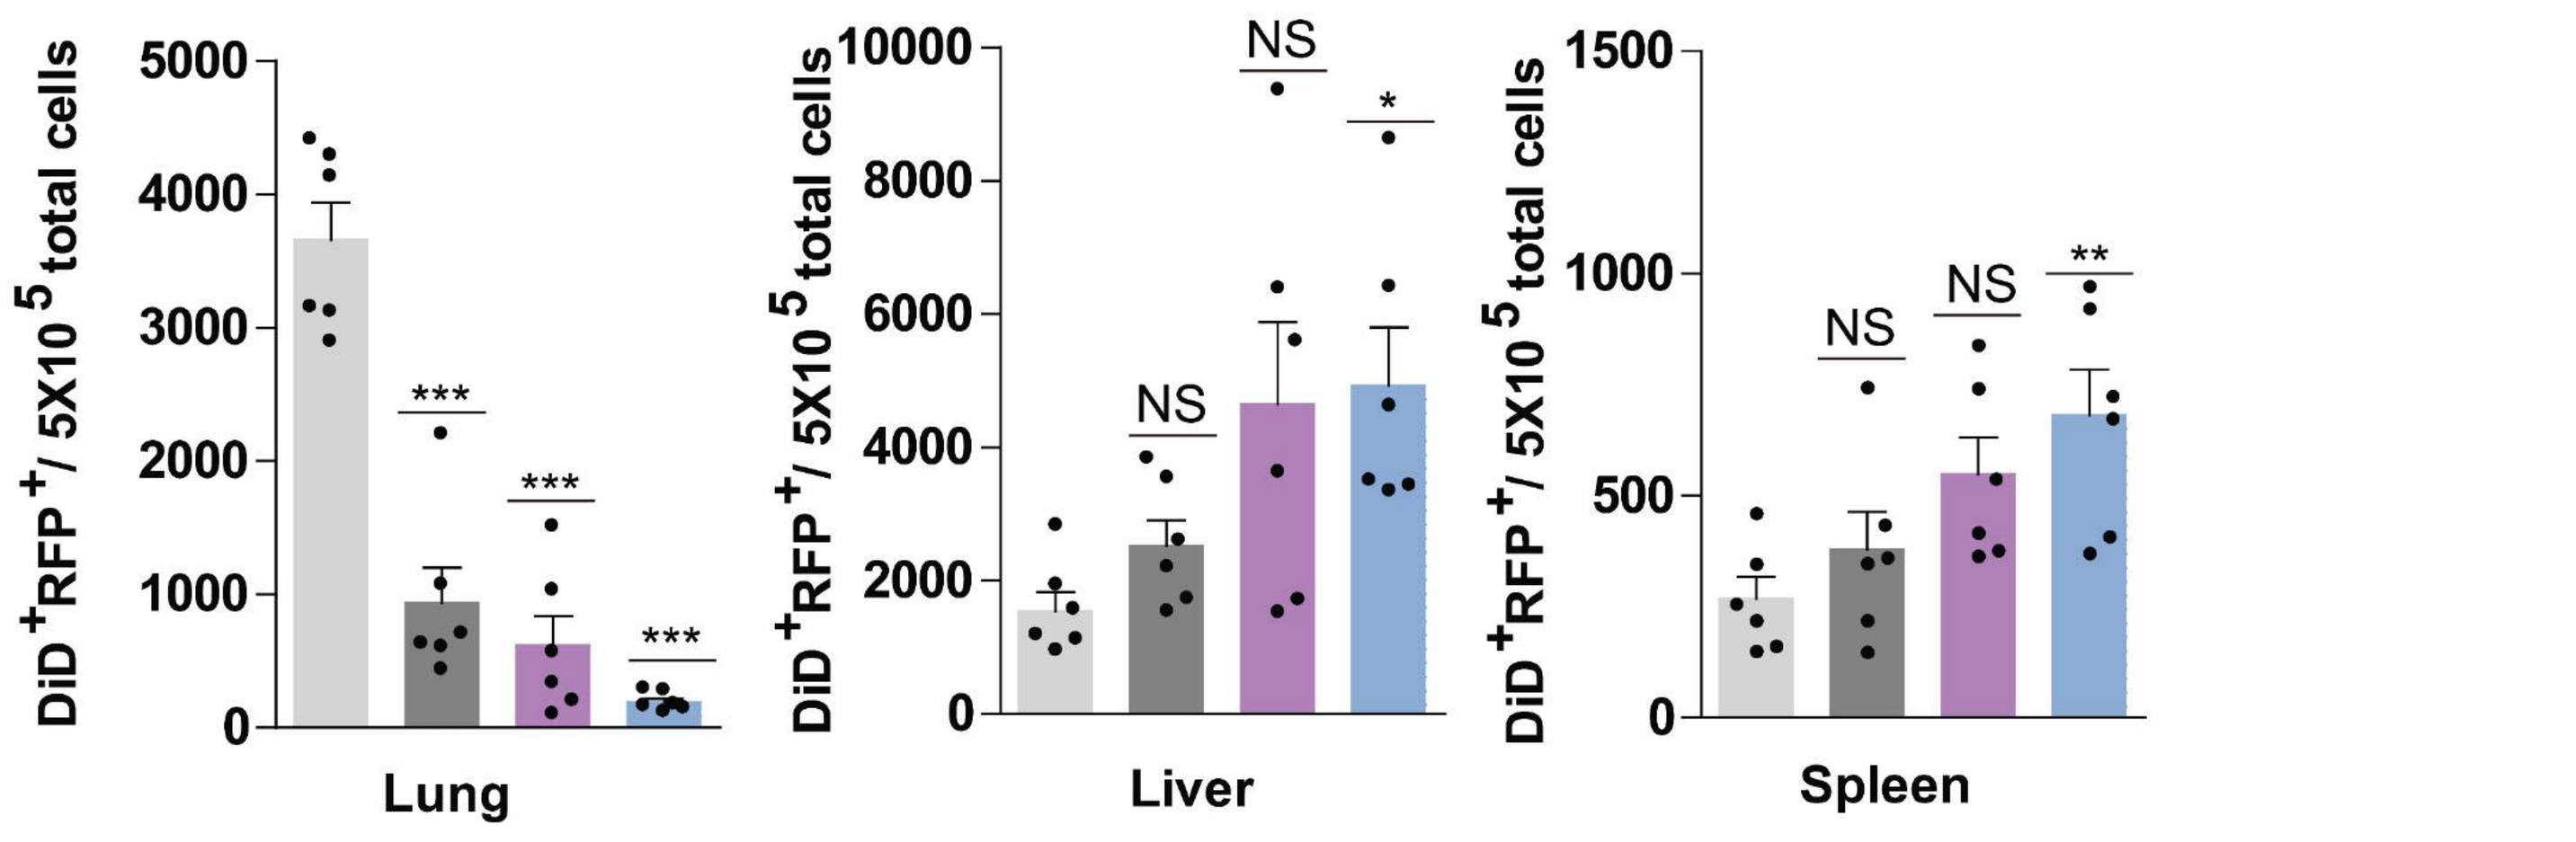
DiD⁺RFP⁺/ 5X10⁵ total cells
5000
4000
3000
2000
1000
0

Lung
10000
8000
6000
4000
2000
0
NS
NS
*
Liver
1500
1000
500
0
NS
NS
**
Spleen

LifeAct RFP MSC were grown in either 2D culture or 3D culture, and then centrifuged in Ficoll to generate 2D or 3D-derived cargocytes. Cells and cargocytes were then washed and stained with DiD intravital dye. One million cells or cargocytes in 2D or 3D were injected retro-orbitally into BALB/c mice. 24 hours after injection, organs were harvested for FACS analysis. Number of DiD stained, RFP+ cells per 5x10^5 events . NS= not significant; * = p<0.05, **=p< 0.01, *** = p<0.001.

Figure 4. 3D cargocytes can be engineered to home to inflamed tissue *in vivo*.

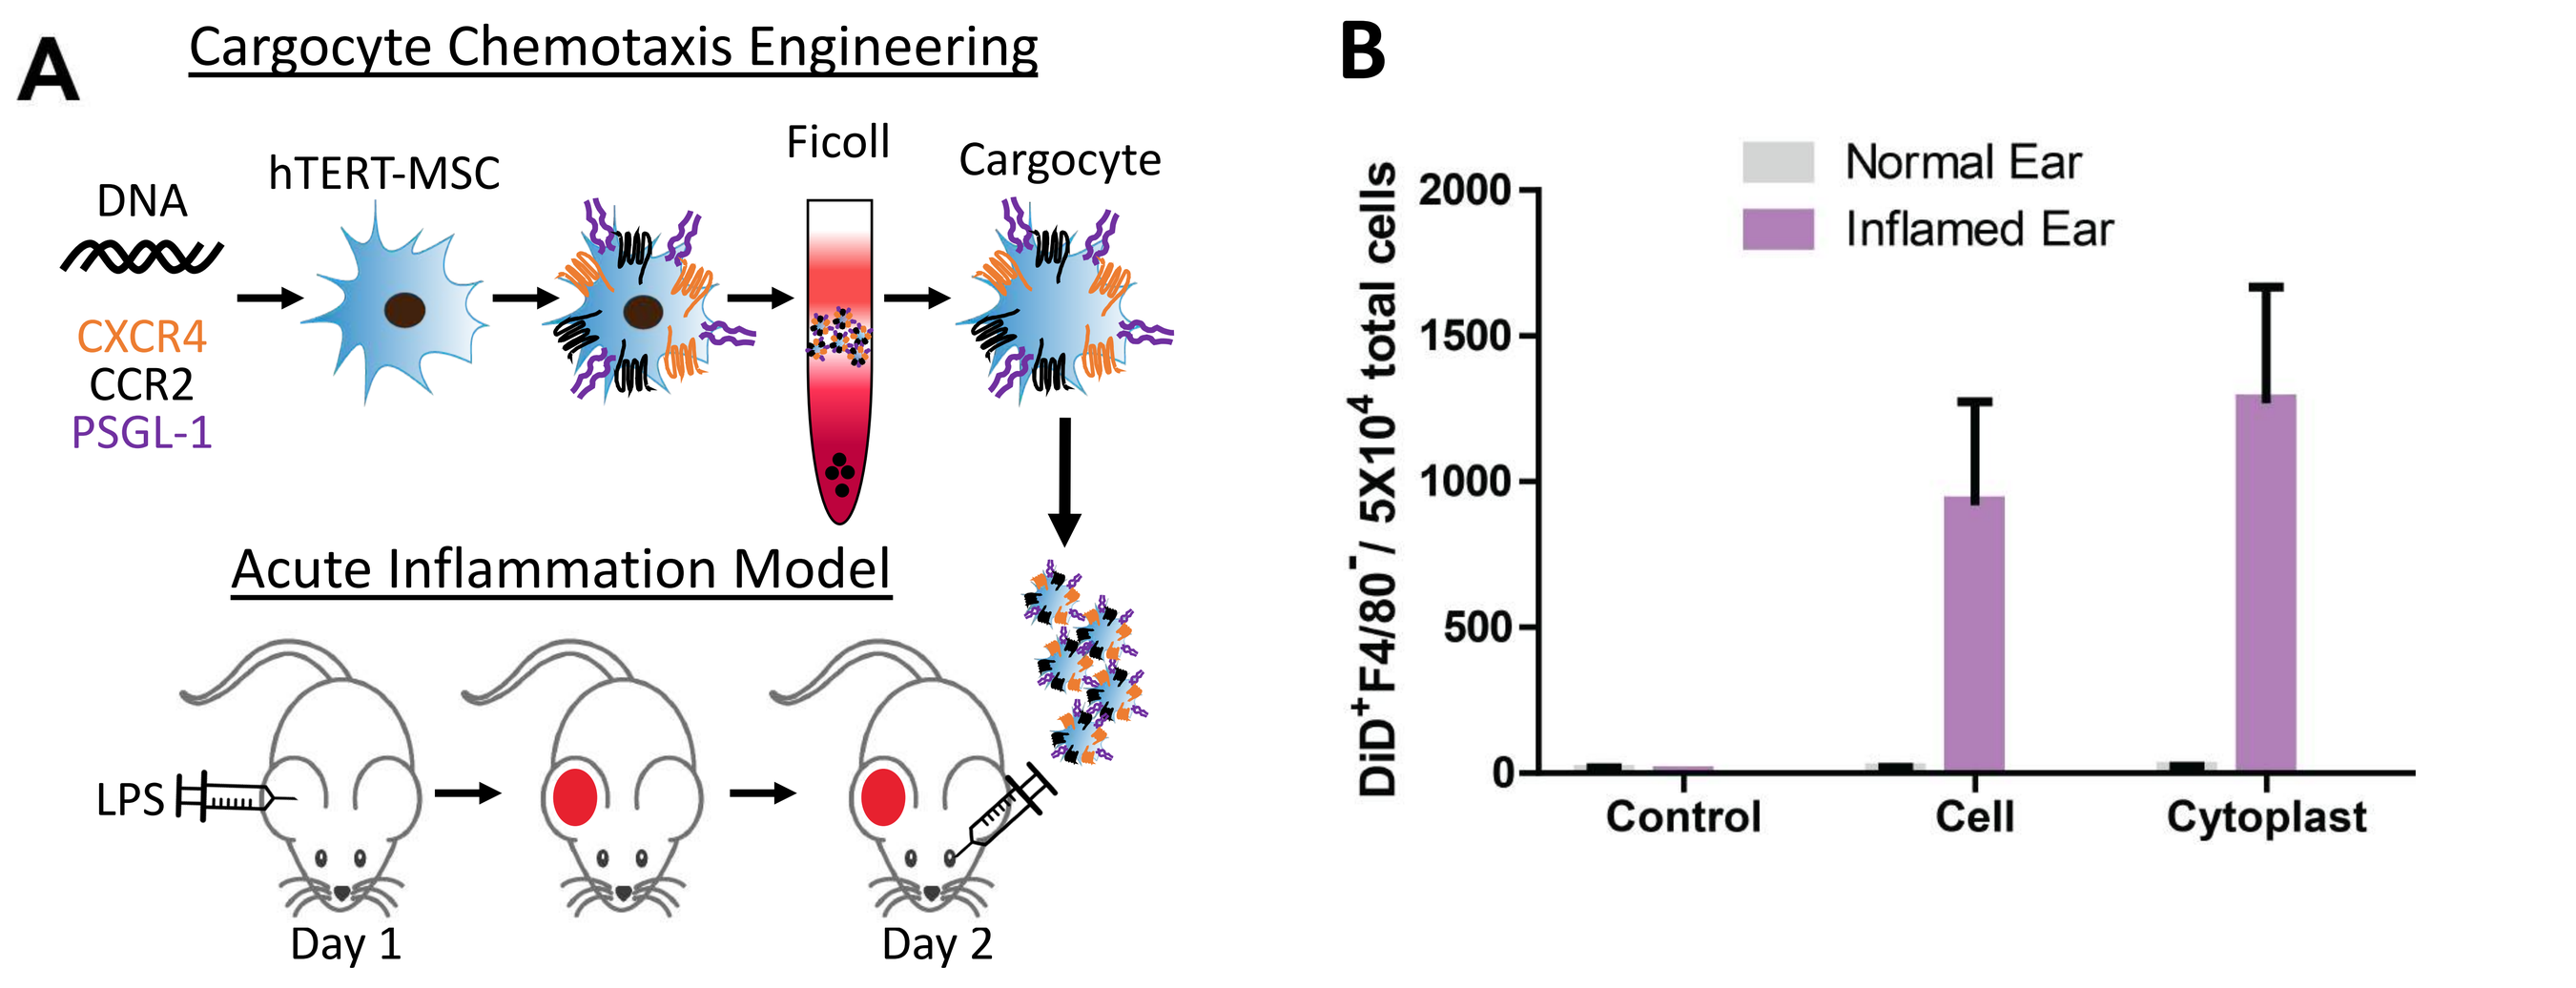
A
Cargocyte Chemotaxis Engineering
DNA
CXCR4
CCR2
PSGL-1
hTERT-MSC
Ficoll
Cargocyte
Acute Inflammation Model
LPS
Day 1
Day 2
B
Normal Ear
Inflamed Ear
DiD⁺F4/80⁻/ 5X10⁴ total cells
2000
1500
1000
500
0
Control
Cell
Cytoplast

A) Schematic of cargocyte engineering for 3 receptors/molecules (triple cargocytes) to improve homing to LPS-induced acute inflammation in the mouse ear. Mice were retro-orbitally injected with 1M 3D-cultured Triple-engineered cargocytes stained with DiD vital dye. B) Cells from mouse ears were analyzed by flow cytometry. Cells from each ear were stained with F4/80 to rule out macrophage phagocytosis. Bar graph quantifies DiD+, F4/80- cargocytes in ears.

Figure 5. 3D-Triple-cargocytes deliver oVSV to micro-metastases.

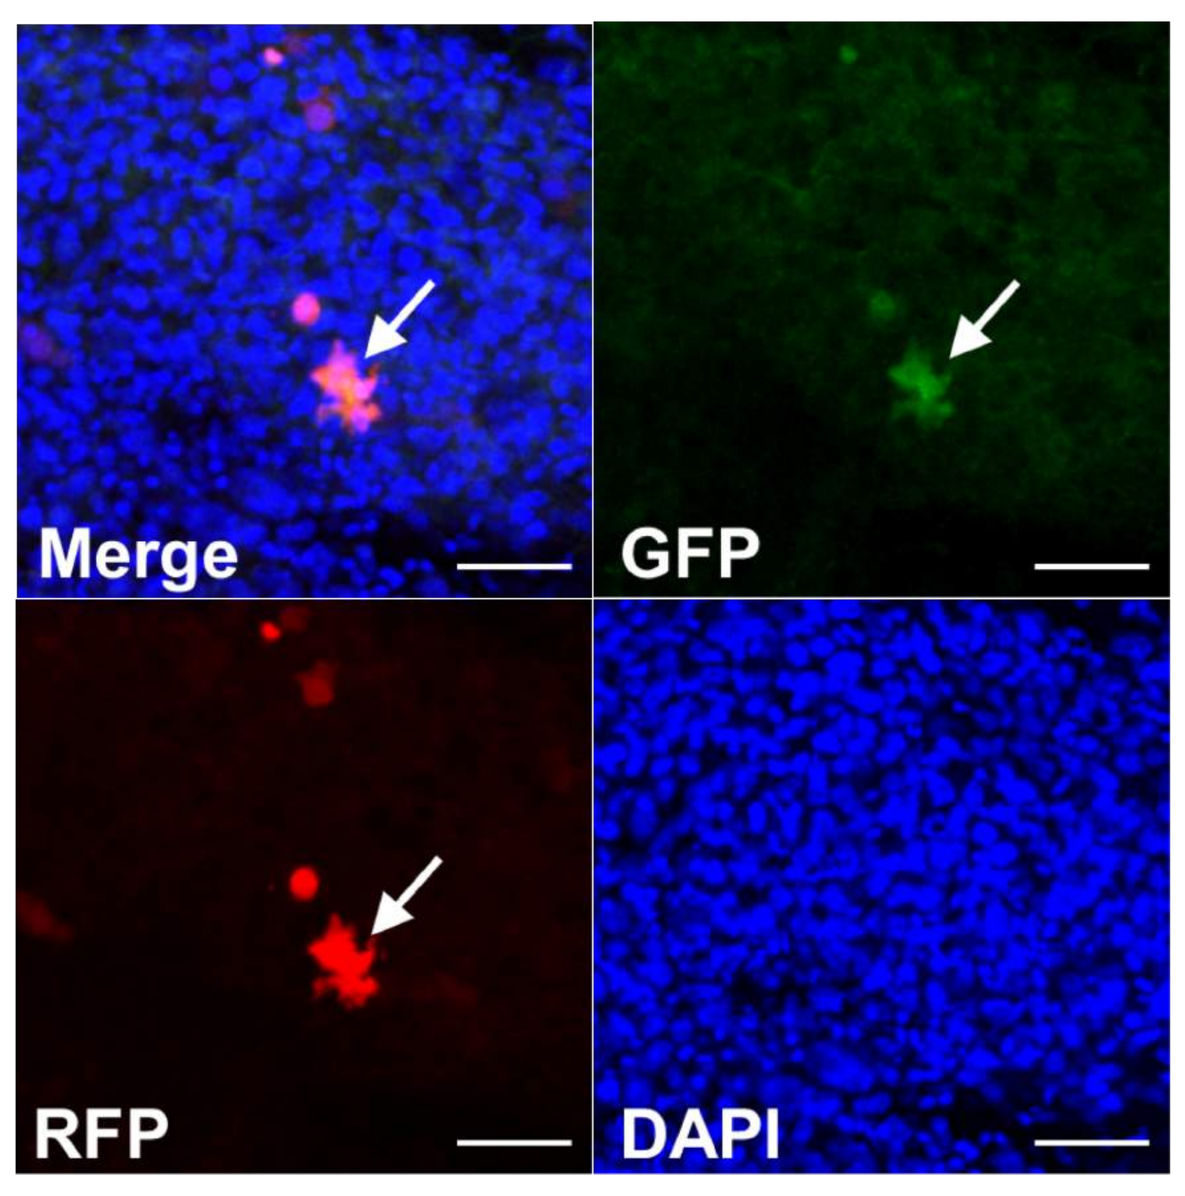
Merge
GFP
RFP
DAPI

Syngeneic metastatic TNBC model was generated in immuno-competent C57Bl/6 mice by retro-orbital injection of 1X10⁶ RFP-labeled E0771 cells. cargocytes infected with 0.5 MOI oncolytic VSV-ΔM51-GFP virus were retro-orbitally injected into the mice 14 days post tumor cell injection. Lung tissues were harvested and fixed 24 hours later. Confocal images were taken with the Olympus FV1000 microscope and analyzed in ImageJ. RFP=E0771 tumors; GFP=VSV-infected cells; Blue=Hoechst 33342-stained cell nuclei. Arrow indicates VSV-infected tumor cells. Scale bar =50 µm.

Figure 6. Strategy to prolong virus persistence in vivo.

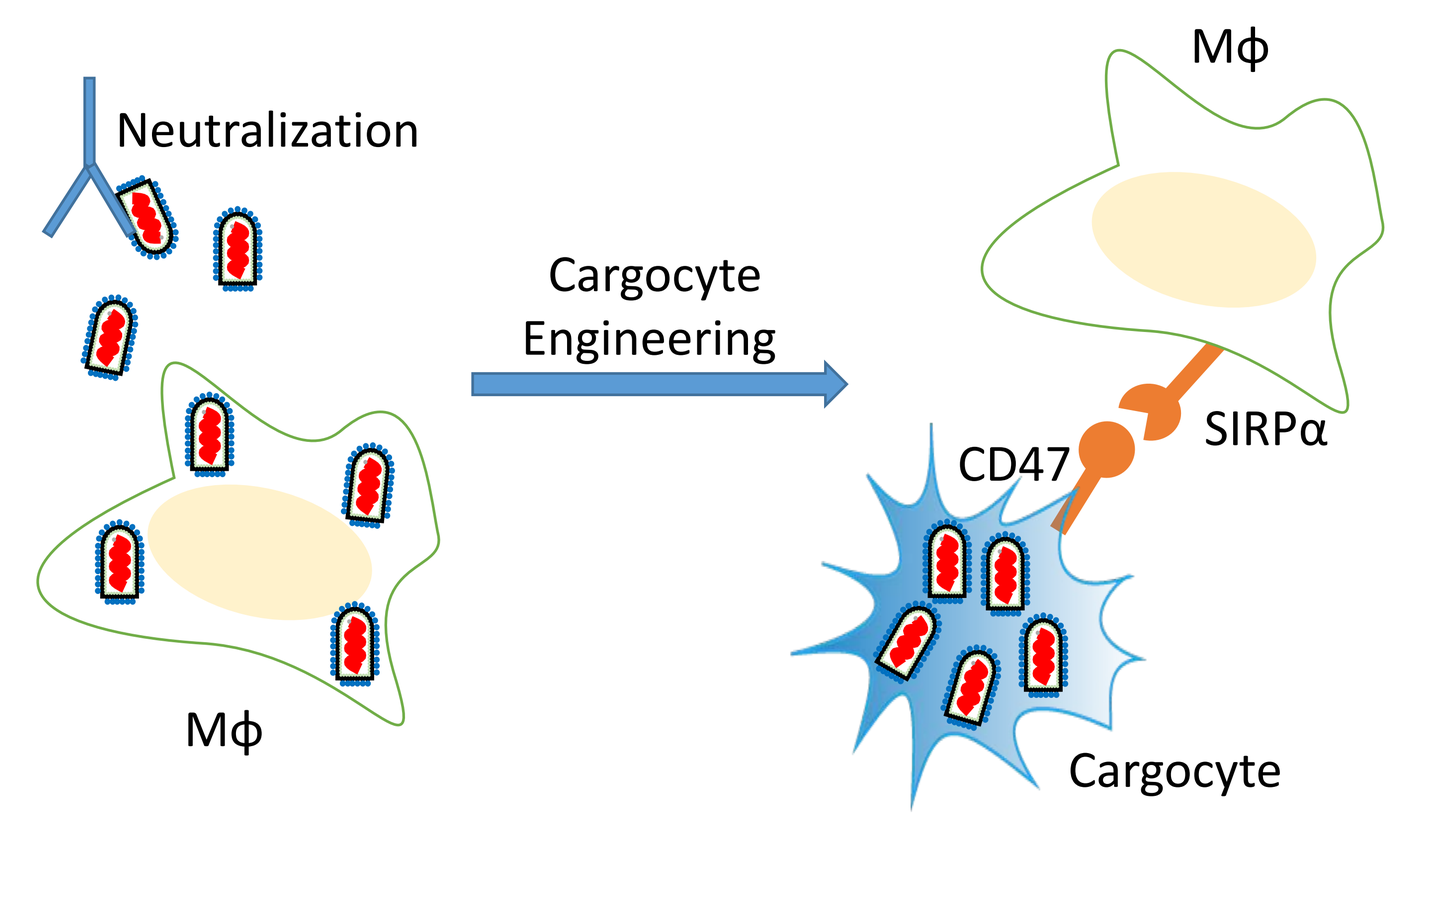
Neutralization
Mϕ
Cargocyte Engineering
Mϕ
CD47
SIRPα
Cargocyte

Cargocyte can be engineered to express CD 47, a "don't eat me" signal, to escape from macrophage or host's mononuclear phagocyte system for systemic delivery.

Conclusions

➢**MSC-derived cargocytes can be effectively loaded with oncolytic viruses**

➢**oVSVs extensively propagate in cargocytes and can be transferred to cancer cells.**

➢**Cargocytes can be extensively engineered to home to inflamed tissues, including tumors *in vivo*.**

➢**Cargocytes are a novel delivery platform for oncolytic viruses, which may improve clinically-relevant delivery and efficacy of oncolytic virus-mediated cancer therapies.**

Acknowledgements: Work is supported by R.L.K. NIH Grants R01 CA097022, CA184594 and CA182495; C.N.A. was supported by NIH 5T32 OD17863-4.

Conflict of Interest: R.L.K. is a founder and CSO of Cytonus Therapeutics, Inc.

References

1. Dent, R. et al. Triple-negative breast cancer: clinical features and patterns of recurrence. Clin Cancer Res (2007) 13, 4429-4434.
2. Kaufman, H. L., Kohlhapp, F. J. & Zloza, A. Oncolytic viruses: a new class of immunotherapy drugs. Nat Rev Drug Discov (2015) 14, 642-662.
3. Fountzilas, C., Patel, S. & Mahalingam, D. Review: Oncolytic virotherapy, updates and future directions. Oncotarget (2017) 8, 102617-102639.
4. Howells, A., Marelli, G., Lemoine, N. R. & Wang, Y. Oncolytic Viruses-Interaction of Virus and Tumor Cells in the Battle to Eliminate Cancer. Front Oncol (2017) 7, 195.
5. Chulpanova, D. S. et al. Application of Mesenchymal Stem Cells for Therapeutic Agent Delivery in Anti-tumor Treatment. Front Pharmacol (2018) 9, 259.
6. Nakashima, H., Kaur, B. & Chiocca, E. A. Directing systemic oncolytic viral delivery to tumors via carrier cells. Cytokine Growth Factor Rev (2010) 21, 119-126.